1086P

Safety of the Natural Killer Cell–Targeted Anti–Killer Cell Immunoglobulin-Like Receptor Antibody Lirilumab in Combination With Nivolumab or Ipilimumab in Two Phase 1 Studies in Advanced Refractory Solid Tumors

Neil H. Segal,[1] Jeffrey Infante,[2] Rachel E. Sanborn,[3] Geoffrey T. Gibney,[4] Donald P. Lawrence,[5] Naiyer Rizvi,[1] Rom Leidner,[3] Tom Gajewski,[6] Erin Bertino,[7] William Sharfman,[8] Sarah Cooley,[9] Suzanne L. Topalian,[8] Walter Urba,[3] Jedd D. Wolchok,[1] Xuemin Gu,[10] Chaitali Passey,[10] Dan McDonald,[10] Praveen Aanur,[10] Shivani Srivastava,[10] F. Stephen Hodi[11]

[1]Memorial Sloan Kettering Cancer Center, New York, NY, USA; [2]Sarah Cannon Research Institute/Tennessee Oncology, PLLC, Nashville, TN, USA; [3]Earle A. Chiles Research Institute, Providence Cancer Center, Portland, OR, USA; [4]Georgetown Lombardi Comprehensive Cancer Center, Washington, DC, USA; [5]Massachusetts General Hospital, Boston, MA, USA; [6]University of Chicago Medical Center; Chicago, IL, USA; [7]The Ohio State University, Columbus, OH, USA; [8]The Sidney Kimmel Comprehensive Cancer Center at Johns Hopkins, Baltimore, MD, USA; [9]University of Minnesota Masonic Cancer Center, Minneapolis, MN, USA; [10]Bristol-Myers Squibb, Princeton, NJ, USA; [11]Dana-Farber Cancer Institute, Boston, MA, USA

Background

- Natural killer (NK) cells play a vital role in the innate immune system's ability to combat viral infections and cancers[1]
 - Activation of NK cells, which is regulated by the balance of positive and negative receptor signaling, results in release of granules containing perforin and granzymes into target cells to induce apoptosis, as well as release of cytokines and chemokines into the micro-environment to recruit other immune cells[1,2]
- The killer cell immunoglobulin-like receptors (KIRs) are a family of inhibitory and activating receptors that are predominantly expressed on NK cells as well as some $CD8^+$ T cells[1-4]
- The binding of inhibitory KIRs to their human leukocyte antigen (HLA) ligands maintains the NK cells in a quiescent state[1-4]
- Therefore, suppression of inhibitory KIR signaling could promote NK-cell activation and antitumor activity[2,5]
- Lirilumab, a fully human IgG4 monoclonal antibody, targets inhibitory KIRs (KIR2DL-1, -2, and -3) to block their interaction with HLA-C and promote antitumor activity of NK cells (**Figure 1**)[2]

Figure 1. Mechanism of Action of Lirilumab

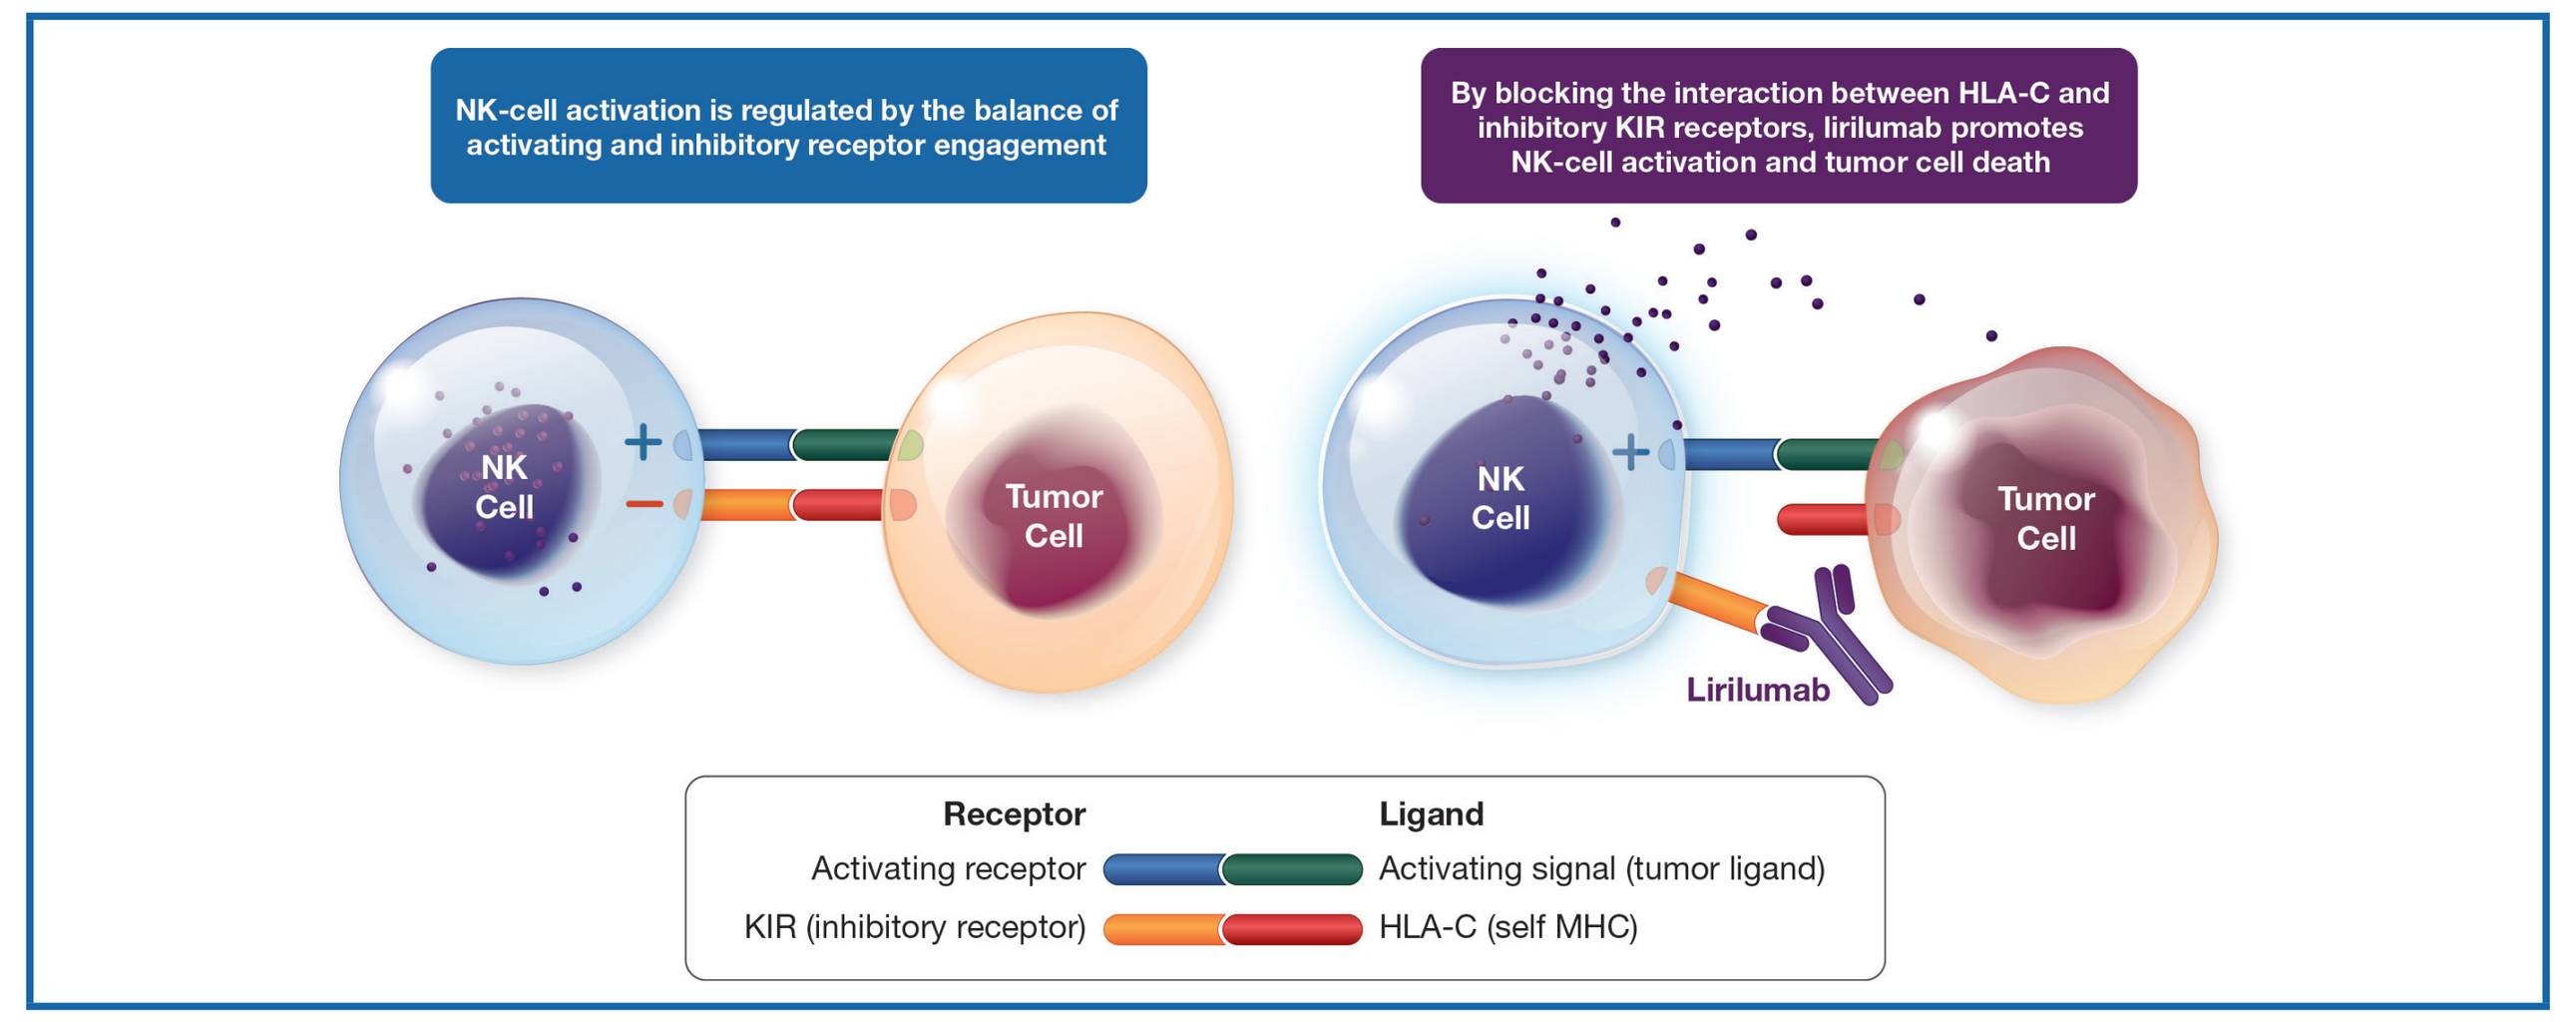

NK-cell activation is regulated by the balance of activating and inhibitory receptor engagement
By blocking the interaction between HLA-C and inhibitory KIR receptors, lirilumab promotes NK-cell activation and tumor cell death
NK Cell
Tumor Cell
NK Cell
Tumor Cell
Lirilumab
Receptor
Ligand
Activating receptor
Activating signal (tumor ligand)
KIR (inhibitory receptor)
HLA-C (self MHC)

HLA-C, human leukocyte antigen-C; KIR, killer cell immunoglobulin-like receptor; MHC, major histocompatibility complex; NK, natural killer.

- The tolerability of lirilumab monotherapy was previously demonstrated in a phase 1 dose-escalation study in patients with solid or hematologic malignancies[6]
- We hypothesized that blocking inhibitory KIR function may potentiate an anti–tumor immune response and complement other immuno-oncology therapies that enhance T-cell activity
- Nivolumab (fully human IgG4 monoclonal antibody inhibitor of programmed death-1 [PD-1] receptor) and ipilimumab (fully human IgG1 monoclonal antibody inhibitor of cytotoxic T-lymphocyte antigen-4 [CTLA-4]) promote antitumor activity of the adaptive immune system through inhibition of immune checkpoint pathways[7-9]
- Two phase 1 studies were initiated to assess the safety, tolerability, and preliminary antitumor activity of lirilumab in combination with nivolumab (CA223-001; NCT01714739) or ipilimumab (CA223-002; NCT01750580) in patients with advanced solid tumors
- Reported here are safety data from the dose-escalation and cohort-expansion phases of CA223-001 (data analysis, August 30, 2016) and the dose-escalation phase of CA223-002 (data analysis, April 20, 2015)

Methods

Study Designs

- CA223-001 is a phase 1 dose-escalation and cohort-expansion study of lirilumab + nivolumab in patients with advanced solid tumors (**Figure 2**)
 - Patients received study therapy in 8-week treatment cycles for a maximum of 12 cycles
 - The dose-escalation phase used a 3 + 3 + 3 design to enroll patients into each dose group to determine the dose-limiting toxicities (DLTs) and maximum tolerated dose (MTD)/maximum administered dose (MAD)
 - Patients received lirilumab 0.1, 0.3, 1.0, or 3.0 mg/kg every 4 weeks + nivolumab 3 mg/kg every 2 weeks
 - After determining the MTD/MAD, an additional 3–12 patients could be enrolled into each dose group to a maximum of 15 patients
 - Cohort expansion was initiated in patients with a range of solid tumors at the MTD (if reached) or MAD

Figure 2. Study Design for CA223-001

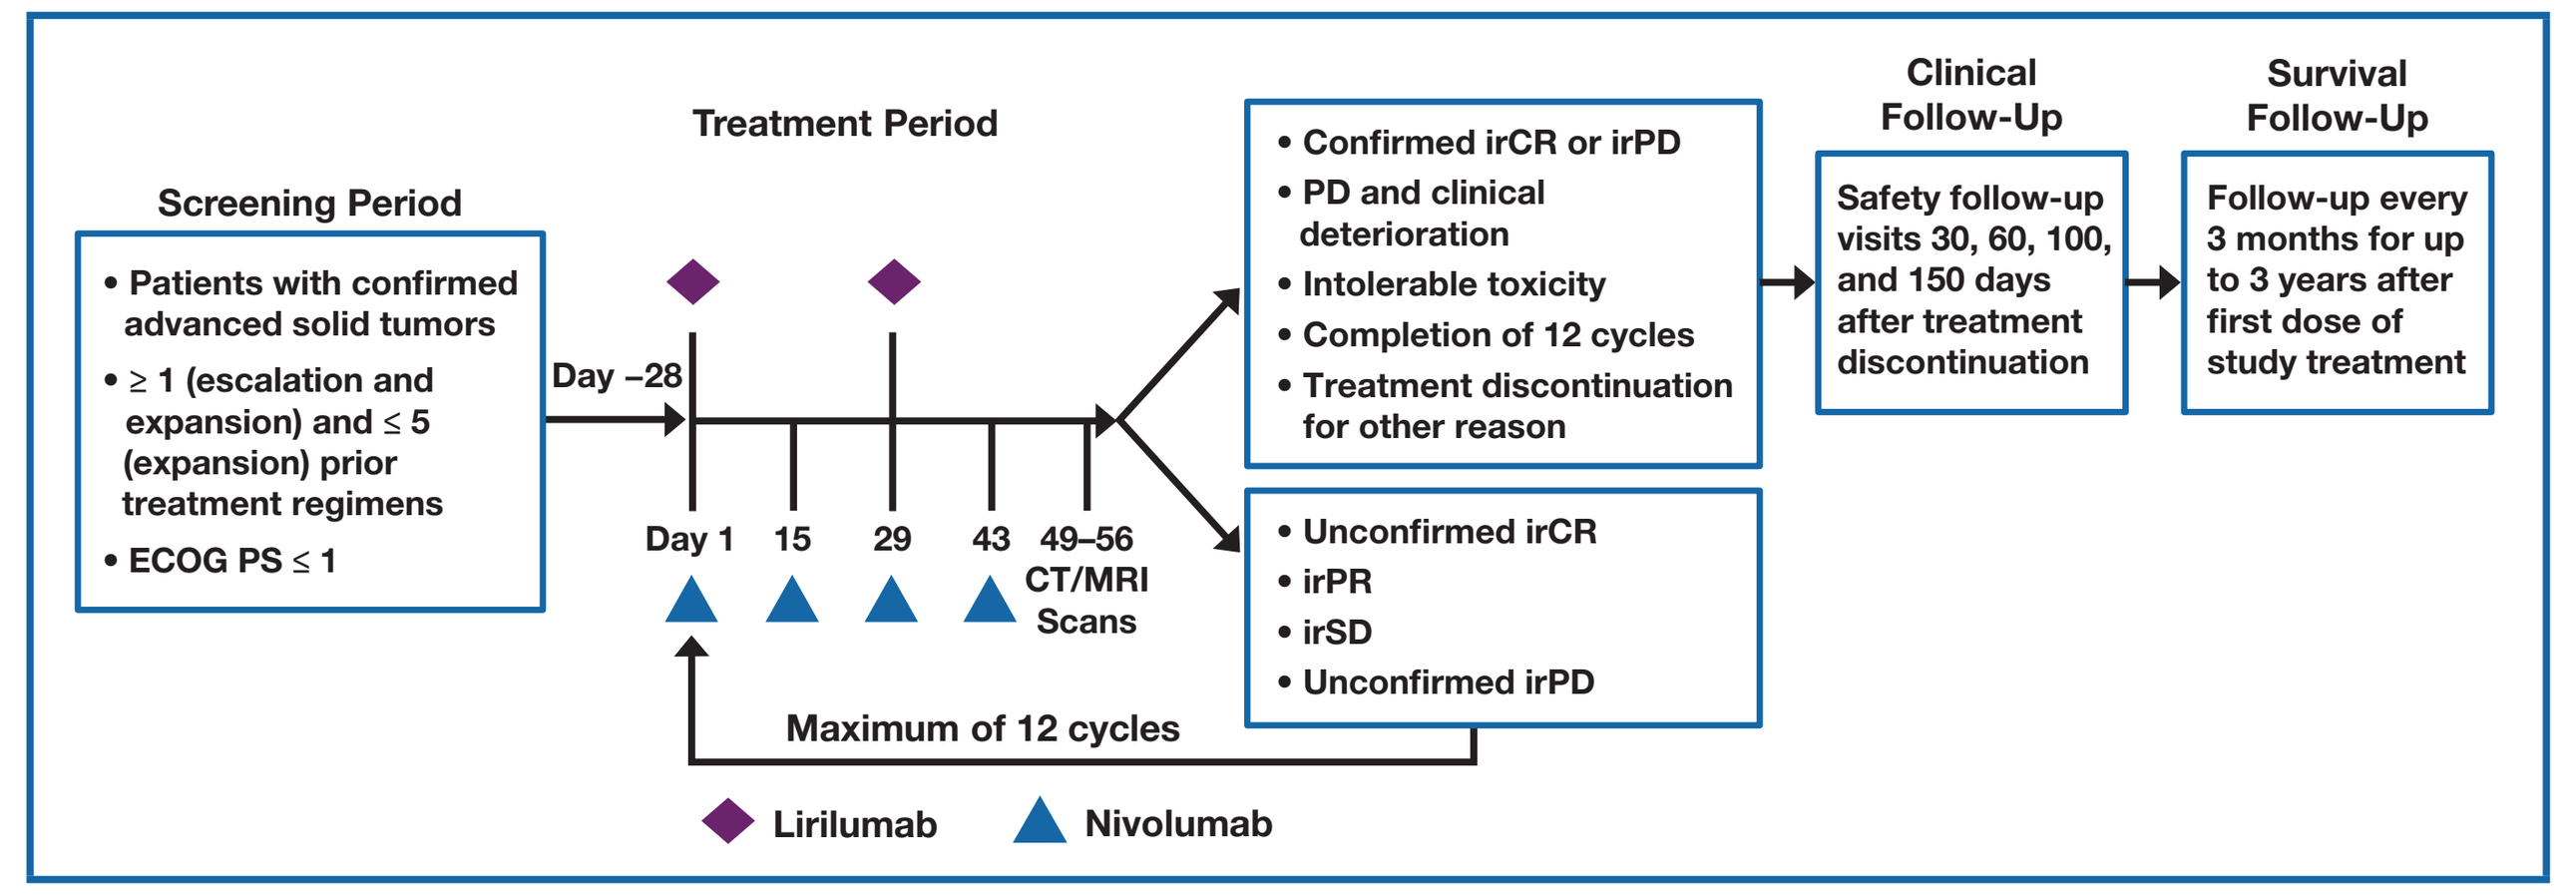

Screening Period
• Patients with confirmed advanced solid tumors
• ≥ 1 (escalation and expansion) and ≤ 5 (expansion) prior treatment regimens
• ECOG PS ≤ 1
Day –28
Treatment Period
Day 1 15 29 43 49–56
CT/MRI Scans
Maximum of 12 cycles
• Confirmed irCR or irPD
• PD and clinical deterioration
• Intolerable toxicity
• Completion of 12 cycles
• Treatment discontinuation for other reason
• Unconfirmed irCR
• irPR
• irSD
• Unconfirmed irPD
Clinical Follow-Up
Safety follow-up visits 30, 60, 100, and 150 days after treatment discontinuation
Survival Follow-Up
Follow-up every 3 months for up to 3 years after first dose of study treatment
Lirilumab
Nivolumab

CT, computed tomography; ECOG PS, Eastern Cooperative Oncology Group performance status; irCR, immune-related complete response; irPD, immune-related progressive disease; irPR, immune-related partial response; irSD, immune-related stable disease; MRI, magnetic resonance imaging; PD, progressive disease.

- CA223-002 was a phase 1 dose-escalation and cohort-expansion study of lirilumab + ipilimumab in patients with advanced solid tumors (**Figure 3**)
 - A 3 + 3 + 3 design was used to enroll patients into the dose-escalation phase to determine the MTD/MAD
 - Patients were to receive lirilumab 0.1, 0.3, 1.0, or 3.0 mg/kg + ipilimumab 3 mg/kg or lirilumab 3.0 mg + ipilimumab 10 mg/kg every 3 weeks for four doses (induction phase) and then every 12 weeks for four doses (maintenance phase)
 - After determining the MTD/MAD, an additional 3–9 patients could be enrolled into each dose group to a maximum of 12 patients
 - The cohort-expansion phase was not initiated for reasons other than safety and tolerability

Figure 3. Study Design for CA223-002

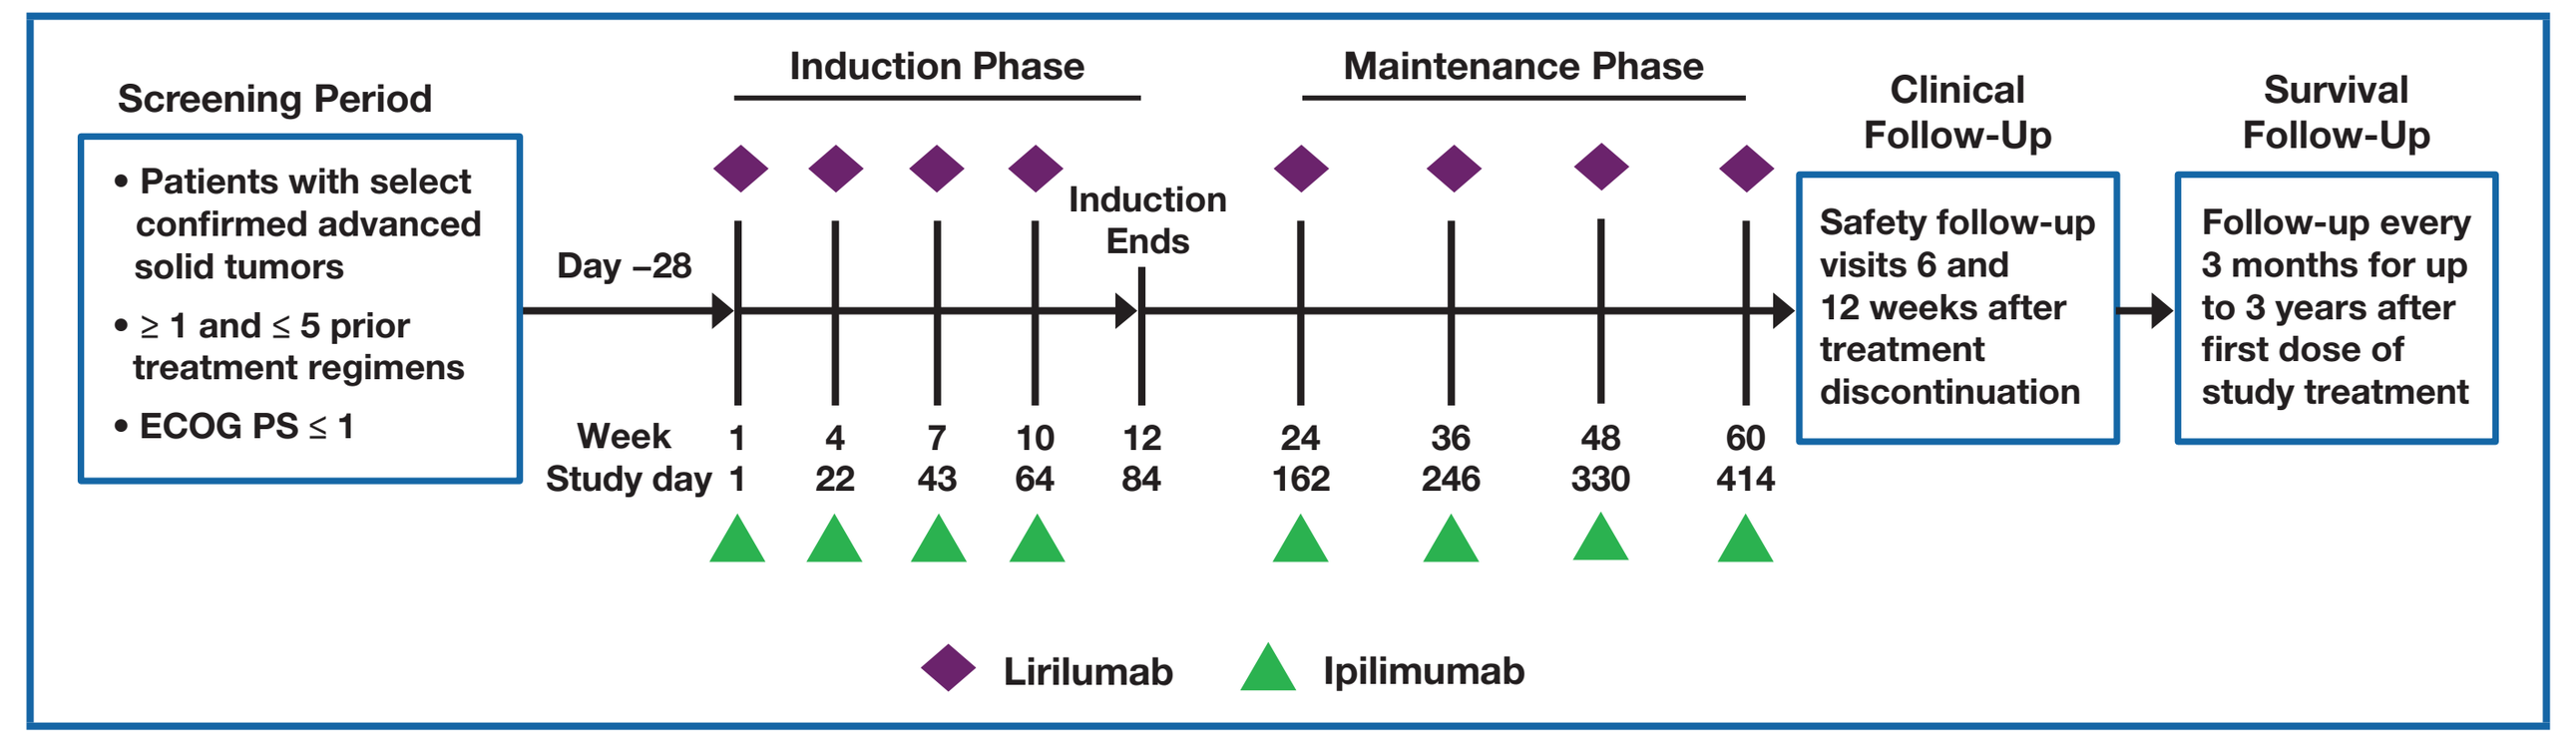

Screening Period
• Patients with select confirmed advanced solid tumors
• ≥ 1 and ≤ 5 prior treatment regimens
• ECOG PS ≤ 1
Day –28
Induction Phase
Maintenance Phase
Induction Ends
Week 1 4 7 10 12 24 36 48 60
Study day 1 22 43 64 84 162 246 330 414
Clinical Follow-Up
Safety follow-up visits 6 and 12 weeks after treatment discontinuation
Survival Follow-Up
Follow-up every 3 months for up to 3 years after first dose of study treatment
Lirilumab
Ipilimumab

ECOG PS, Eastern Cooperative Oncology Group performance status.

- In CA223-001 and CA223-002, the decision to treat a patient with additional cycles of therapy was made based on tumor assessments performed at the end of each cycle and before the first dose of the next cycle according to immune-related Response Evaluation Criteria In Solid Tumors[10]
 - Patients were treated until the first occurrence of immune-related complete response or progressive disease, completion of the maximum number of cycles, clinical deterioration suggesting that no further benefit was likely, intolerability of treatment, or discontinuation due to other reason

Primary Endpoints (CA223-001 and CA223-002)

- To assess the safety and tolerability and to identify the DLTs and MTD of study therapy
- To assess the preliminary antitumor activity of lirilumab + nivolumab (CA223-001 only)

Secondary Endpoints (CA223-001 and CA223-002)

- To evaluate the antitumor activity (CA223-002 only), pharmacokinetics, pharmacodynamics, and immunogenicity of lirilumab when administered with either nivolumab or ipilimumab

Key Inclusion Criteria

- Eligible patients were ≥ 18 years old with confirmed advanced solid tumors who had progressed on or were intolerant of ≥ 1 (both studies) and ≤ 5 (CA223-001 cohort expansion; CA223-002) prior treatment regimens
 - In CA223-001, patients with all solid tumors (except central nervous system tumors) were eligible for dose escalation, and those with select advanced solid tumors were eligible for the cohort-expansion phase
 - In CA223-002, eligible patients had select advanced solid tumors
- Patients were required to have an Eastern Cooperative Oncology Group performance status ≤ 1 (both studies)[11]

Key Exclusion Criteria

- Patients with active or prior history of autoimmune disease or who received prior treatment with any of the following were not eligible:
 - Anti–KIR antibody (both studies)
 - Anti–PD-1 or anti–programmed death-1 ligand 1 (anti–PD-L1) antibody (CA223-001 only)
 - Anti–CTLA-4 antibody; however, in CA223-001, prior anti–CTLA-4 treatment that occurred > 100 days before the start of study treatment was permitted
 - Other immune-modulating antibody (both studies)
- Patients were not permitted to have received prior therapy within 28 days of starting study treatment (both studies)

Safety Assessments

- Adverse events (AEs) were assessed continually during treatment and for 150 days (CA223-001) and 90 days (CA223-002) following the last dose per Common Terminology Criteria for Adverse Events v4.0[12]
- DLTs were assessed during the first 8 (CA223-001) and 9 (CA223-002) weeks of treatment

Results from CA223-001

Patients

- As of August 30, 2016, 159 patients were treated in the escalation and expansion phases
 - Patients received nivolumab 3.0 mg/kg and lirilumab 0.1 (n = 4), 0.3 (n = 16), 1.0 (n = 15), or 3.0 (n = 124) mg/kg
- Baseline patient characteristics are described in **Table 1**
- Approximately 75% of patients had discontinued treatment at the time of data analysis (**Table 2**)

Table 1. Baseline Patient Characteristics in CA223-001

	CA223-001 All Patients (N = 159)
Mean age (range), years	58.5 (21–85)
Median age, years	60.0
Age < 65 years, n (%)	99 (62.3)
Male, n (%)	98 (61.6)
Race, n (%)	
White	139 (87.4)
Black	7 (4.4)
Native American/Alaska native	1 (0.6)
Asian	8 (5.0)
Other	4 (2.5)

Table 2. Patient Disposition in CA223-001

	CA223-001 All Patients (N = 159)
Continuing treatment, n (%)	43 (27.0)
Not continuing treatment, n (%)	116 (73.0)
Reasons for not continuing treatment, n (%)	
Disease progression	84 (52.8)
Study drug toxicity	8 (5.0)
Patient decision	6 (3.8)
AE unrelated to study drug	4 (2.5)
Patient withdrew consent	4 (2.5)
Maximum clinical benefit	3 (1.9)
Patient no longer met study criteria	2 (1.3)
Lost to follow-up	1 (0.6)
Noncompliance	1 (0.6)
Other	3 (1.9)

AE, adverse event.

Safety and Tolerability

- No DLTs were reported with lirilumab + nivolumab treatment
 - The MTD was not determined; therefore, cohort expansion was initiated at the MAD (lirilumab 3.0 mg/kg + nivolumab 3.0 mg/kg)
- Across treatment groups, the median duration of treatment with lirilumab and nivolumab ranged from ≈ 14–58 weeks
- Most patients (71.7%) experienced at least one treatment-related AE (TRAE; **Table 3**)
 - TRAEs were generally grade 1 or 2, and the most common TRAEs (≥ 10%) among all treated patients were fatigue (20.8%), pruritus (18.9%), infusion-related reaction (17.6%), and rash (16.4%)
- Infusion-related reactions (often associated with lirilumab monotherapy) occurred in 28 patients
 - These TRAEs were grade 1 or 2 and primarily occurred after the first infusion; pretreatment was not needed

Table 3. All TRAEs and TRAEs Reported in ≥ 5% of All Patients in CA223-001

Patients With a TRAE, n (%)	Lirilumab 0.1 mg/kg + Nivolumab 3.0 mg/kg n = 4		Lirilumab 0.3 mg/kg + Nivolumab 3.0 mg/kg n = 16		Lirilumab 1.0 mg/kg + Nivolumab 3.0 mg/kg n = 15		Lirilumab 3.0 mg/kg + Nivolumab 3.0 mg/kg n = 124		All Patients N = 159	
	Any Grade	Grade 3/4[a]	Any Grade	Grade 3/4[b]	Any Grade	Grade 3/4[c]	Any Grade	Grade 3/4[d]	Any Grade	Grade 3/4[a-d]
Any TRAE	4 (100)	2 (50.0)	15 (93.8)	2 (12.5)	14 (93.3)	3 (20.0)	81 (65.3)	17 (13.7)	114 (71.7)	24 (15.1)
TRAEs in ≥ 5% of all patients										
Fatigue	2 (50.0)	0	5 (31.3)	0	4 (26.7)	0	22 (17.7)	0	33 (20.8)	0
Pruritus	2 (50.0)	0	3 (18.8)	0	6 (40.0)	0	19 (15.3)	0	30 (18.9)	0
Infusion-related reaction	1 (25.0)	0	1 (6.3)	0	7 (46.7)	0	19 (15.3)	0	28 (17.6)	0
Rash	1 (25.0)	0	5 (31.3)	0	4 (26.7)	0	16 (12.9)	0	26 (16.4)	0
Diarrhea	0	0	0	0	3 (20.0)	1 (6.7)	10 (8.1)	0	13 (8.2)	1 (0.6)
Arthralgia	0	0	3 (18.8)	0	4 (26.7)	0	6 (4.8)	0	13 (8.2)	0
Amylase increased	0	0	2 (12.5)	0	1 (6.7)	0	9 (7.3)	4 (3.2)	12 (7.5)	4 (2.5)
Maculopapular rash	0	0	2 (12.5)	0	1 (6.7)	1 (6.7)	9 (7.3)	1 (0.8)	12 (7.5)	2 (1.3)
Nausea	1 (25.0)	0	2 (12.5)	0	2 (13.3)	0	7 (5.6)	0	12 (7.5)	0
Appetite decreased	2 (50.0)	0	0	0	1 (6.7)	0	6 (4.8)	0	9 (5.7)	0
Pyrexia	0	0	0	0	0	0	8 (6.5)	0	8 (5.0)	0

AST, aspartate aminotransferase; TRAE, treatment-related adverse event.
[a]Grade 3/4 TRAEs included hypophosphatemia and QT interval prolonged (n = 1 for each); [b]Grade 3/4 TRAEs included leukopenia, neutropenia, and pulmonary embolism (n = 1 for each); [c]Grade 3/4 TRAEs not shown in the table included blood pressure increased (n = 1); [d]Grade 3/4 TRAEs not shown in the table included lipase increased (n = 4) and AST increased, autoimmune pancreatitis, bacteremia, drug hypersensitivity, dyspnea, hyperglycemia, hypertension, hypophosphatemia, hypopituitarism, portal vein thrombosis, radiation skin injury, stridor, and thrombocytopenia (n = 1 for each).

- TRAEs leading to discontinuation were reported in 12 patients (7.5%), with only treatment-related pneumonitis (n = 3; all grade 2) and diarrhea (n = 2; all grade 2) occurring in > 1 patient
- Treatment-related serious AEs (TRSAEs) occurred in 10 patients (6.3%); diarrhea (n = 2; all grade 2) and pneumonitis (n = 2; all grade 2) were the only events experienced by > 1 patient
 - Grade 3 or 4 TRSAEs were experienced by four patients and included autoimmune pancreatitis (asymptomatic), dyspnea, thrombocytopenia, and radiation skin injury
- No treatment-related deaths were reported

Summary of Results from CA223-002

- On April 20, 2015, CA223-002 was terminated for reasons other than safety and tolerability
 - At that time, 22 patients enrolled in the escalation phase had received ipilimumab 3.0 mg/kg and lirilumab 0.1 (n = 3), 0.3 (n = 8), 1.0 (n = 6) or 3.0 (n = 5) mg/kg
- Baseline patient characteristics are described in **Table 4**

Table 4. Baseline Patient Characteristics in CA223-002

	CA223-002 All Patients (N = 22)
Mean age (range), years	58.0 (30–85)
Age < 65 years, n (%)	14 (63.6)
Male, n (%)	12 (54.5)
Race, n (%)	
White	22 (100)

- Median duration of treatment across dose groups ranged from 9–12 (induction phase) and 0–48 (maintenance phase) weeks
- DLTs included grade 2 iridocyclitis (n = 1; lirilumab 0.3 mg/kg + ipilimumab 3.0 mg/kg) and grade 3 rash (n = 1; lirilumab 1.0 mg/kg + ipilimumab 3.0 mg/kg)
 - Grade 2 iridocyclitis was reported as a DLT because it did not resolve to grade 1 within 2 weeks despite treatment; this event was also considered to be related to nivolumab, which the patient received 32 days before starting study therapy
 - This was the only patient to discontinue due to a TRAE
- TRAEs were mostly grade 1 or 2, with fatigue (27.3%) and diarrhea (22.7%) being the most common (**Table 5**)
- TRSAEs occurred in two patients (9.1%; grade 3 hypopituitarism and grade 2 iridocyclitis)
- No treatment-related deaths were reported

Table 5. All TRAEs and TRAEs Reported in ≥ 5% of All Patients in CA223-002

Patients With a TRAE, n (%)	Lirilumab 0.1 mg/kg + Ipilimumab 3.0 mg/kg n = 3		Lirilumab 0.3 mg/kg + Ipilimumab 3.0 mg/kg n = 8		Lirilumab 1.0 mg/kg + Ipilimumab 3.0 mg/kg n = 6		Lirilumab 3.0 mg/kg + Ipilimumab 3.0 mg/kg n = 5		All Patients N = 22	
	Any Grade	Grade 3/4	Any Grade	Grade 3/4[a]	Any Grade	Grade 3/4[b]	Any Grade	Grade 3/4	Any Grade	Grade 3/4[a,b]
Any TRAE	3 (100)	0	6 (75.0)	1 (12.5)	3 (50.0)	1 (16.7)	3 (60.0)	0	15 (68.2)	2 (9.1)
TRAEs in ≥ 5% of all patients										
Fatigue	2 (66.7)	0	1 (12.5)	0	2 (33.3)	0	1 (20.0)	0	6 (27.3)	0
Diarrhea	0	0	2 (25.0)	0	2 (33.3)	0	1 (20.0)	0	5 (22.7)	0
Nausea	1 (33.3)	0	3 (37.5)	0	0	0	0	0	4 (18.2)	0
Appetite decreased	1 (33.3)	0	2 (25.0)	0	1 (16.7)	0	0	0	4 (18.2)	0
Vomiting	0	0	3 (37.5)	0	0	0	1 (20.0)	0	4 (18.2)	0
Chills	2 (66.7)	0	1 (12.5)	0	0	0	1 (20.0)	0	4 (18.2)	0
Rash	0	0	1 (12.5)	0	2 (33.3)	0	0	0	3 (13.6)	0
Pruritic rash	1 (33.3)	0	2 (25.0)	0	0	0	0	0	3 (13.6)	0
Pyrexia	2 (66.7)	0	0	0	0	0	1 (20.0)	0	3 (13.6)	0
Hyperhidrosis	2 (66.7)	0	0	0	0	0	0	0	2 (9.1)	0
Maculopapular rash	0	0	2 (25.0)	0	0	0	0	0	2 (9.1)	0
Headache	1 (33.3)	0	0	0	0	0	1 (20.0)	0	2 (9.1)	0
Pruritus	0	0	0	0	1 (16.7)	1 (16.7)	1 (20.0)	0	2 (9.1)	1 (4.5)
Hypertension	0	0	1 (12.5)	0	1 (16.7)	0	0	0	2 (9.1)	0

TRAE, treatment-related adverse event.
[a]Grade 3/4 TRAEs not shown in the table included hypopituitarism (n = 1); [b]Grade 3/4 TRAEs not shown in the table included erythematous rash (n = 1).

Conclusions

- There does not appear to be additional safety concerns with the lirilumab + ipilimumab combination compared to ipilimumab monotherapy in the limited population studied
- No additional safety concerns were observed with the lirilumab + nivolumab combination therapy compared to nivolumab monotherapy, with the exception of an increase in low-grade infusion-related reactions that were clinically manageable
 - Low-grade infusion-related reactions more commonly associated with lirilumab monotherapy occurred mostly after the first administration of therapy and did not require pretreatment
- Based on these data, further evaluation of lirilumab in combination with nivolumab is warranted
- Efficacy data from the lirilumab + nivolumab combination study will be reported separately and are forthcoming

References

1. Purdy AK, Campbell KS. *Cancer Biol Ther.* 2009;8:2211–20.
2. Kohrt HE et al. *Blood.* 2014;123:678–86.
3. Huard B, Karlsson L. *Nature.* 2000;403:325–8.
4. Uhrberg M et al. *J Immunol.* 2001;166:3923–32.
5. Romagne F et al. *Blood.* 2009;114:2667–77.
6. Vey N et al. *J Clin Oncol.* 2015;32:(15 suppl). Abstract 3065.
7. Wang C et al. *Cancer Immunol Res.* 2014;2:846–56.
8. Opdivo [package insert]. Princeton, NJ: Bristol-Myers Squibb; 2016.
9. Yervoy [package insert]. Princeton, NJ: Bristol-Myers Squibb; 2015.
10. Nishino M et al. *J Immunother Cancer.* 2014;2:17-1426-2-17. eCollection 2014.
11. Oken MM et al. *Am J Clin Oncol.* 1982;5:649–55.
12. Common Terminology Criteria for Adverse Events (CTCAE) version 4.0. National Cancer Institute website. http://evs.nci.nih.gov/ftp1/CTCAE/CTCAE_4.03_2010-06-14_QuickReference_8.5x11.pdf. Revised June 2010. Accessed September 13, 2016.

Acknowledgments

- The patients and families that made this trial possible
- The clinical study teams that participated in this trial
- Bristol-Myers Squibb, Inc., and Ono Pharmaceutical Company Limited, Osaka, Japan
- The study was supported by Bristol-Myers Squibb, Inc.
- All authors contributed to and approved the presentation; writing and editorial assistance was provided by Christopher Reina, of Chrysalis Medical Communications, Inc., funded by Bristol-Myers Squibb

Receive an electronic PDF of this poster on your mobile phone:
– Go to getscanlife.com from your mobile browser to download the free barcode reader application
– Scan the code and get access to content

Presented at the European Society for Medical Oncology Annual Meeting; October 7–11, 2016; Copenhagen, Denmark

Email: segaln@mskcc.org

Copies of this poster obtained through Quick Response (QR) Code are for personal use only and may not be reproduced without written permission of the authors.