Repeated Dosing Of Anti-KIR (IPH2101) As Maintenance Therapy In Elderly Patients With Acute Myeloid Leukemia

Abstract # 2696

IPH2101-102 Trial (NCT01256073)

N Vey[1], JH Bourhis[2], C Recher[3], A Etienne[1], A Charbonnier[1], J Rey[1], P André[4], F Calmels[4], R Zerbib[4], R Buffet[4], T Prebet[1]

1 - Institut Paoli-Calmettes, Marseilles, France; 2 - Institut Gustave Roussy, Villejuif, France; 3 - CHU Purpan, Toulouse, France; 4 –Innate Pharma, Marseille, France

Background

Mechanism of Action: anti-KIR mAbs block an inhibitory signal to NK cells

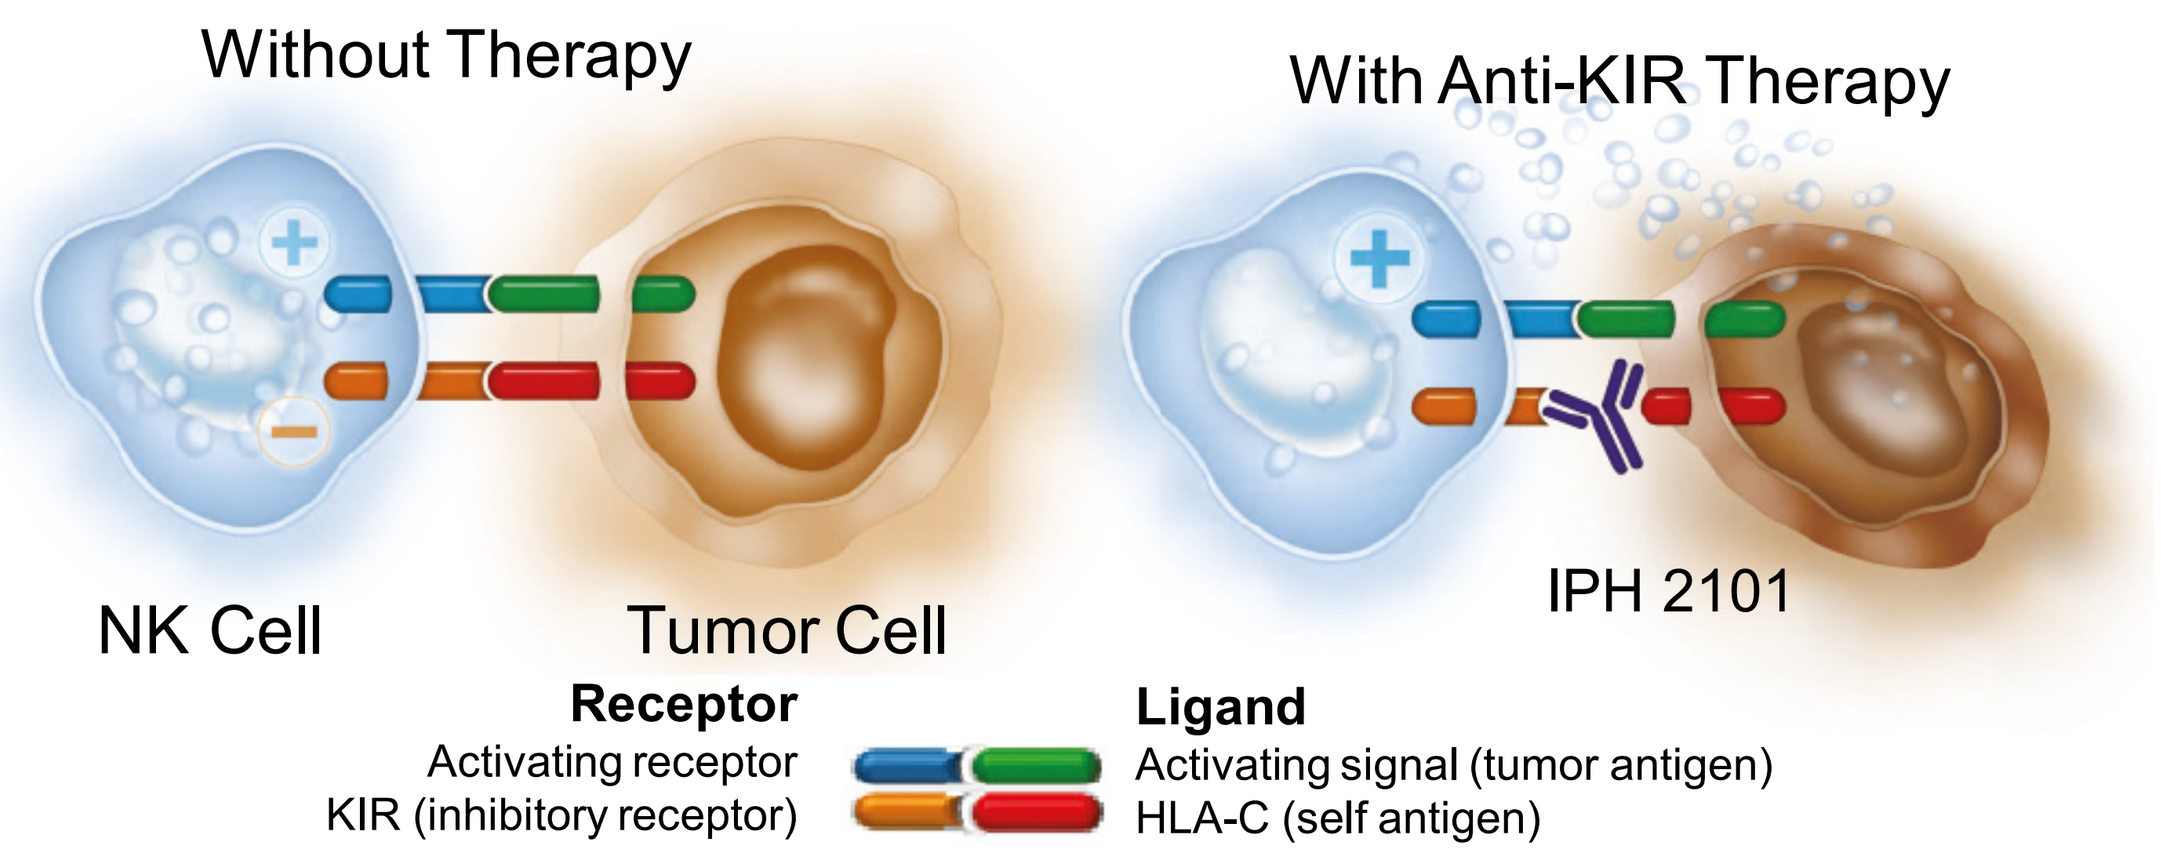
Without Therapy
With Anti-KIR Therapy
NK Cell
Tumor Cell
IPH 2101
Receptor
Activating receptor
KIR (inhibitory receptor)
Ligand
Activating signal (tumor antigen)
HLA-C (self antigen)

- NK cell activation is determined by the balance of activating (positive) and inhibitory (negative) receptor stimulation
- Tumor cells are able to evade innate immunity through the interaction of KIR with HLA-C
- By blocking this interaction, anti-KIR IPH2101 facilitates the activation of NK cells and subsequent apoptosis of the tumor cell

Anti-KIR IPH2101

- IPH2101 is a the first generation of anti-KIR antibody (now succeeded by lirilumab)
- Targets KIR2DL expressed by NK cells (but also targets 2DS)
- Blocks interactions between KIRs and their HLA-C ligands

Preclinical *In vivo* Efficacy of Anti-KIR

- Anti-KIR blockade by IPH2101 (a first generation of anti-KIR antibody) resulted in long-term survival in SCID mice inoculated with lethal autologous AML cells *(1)*

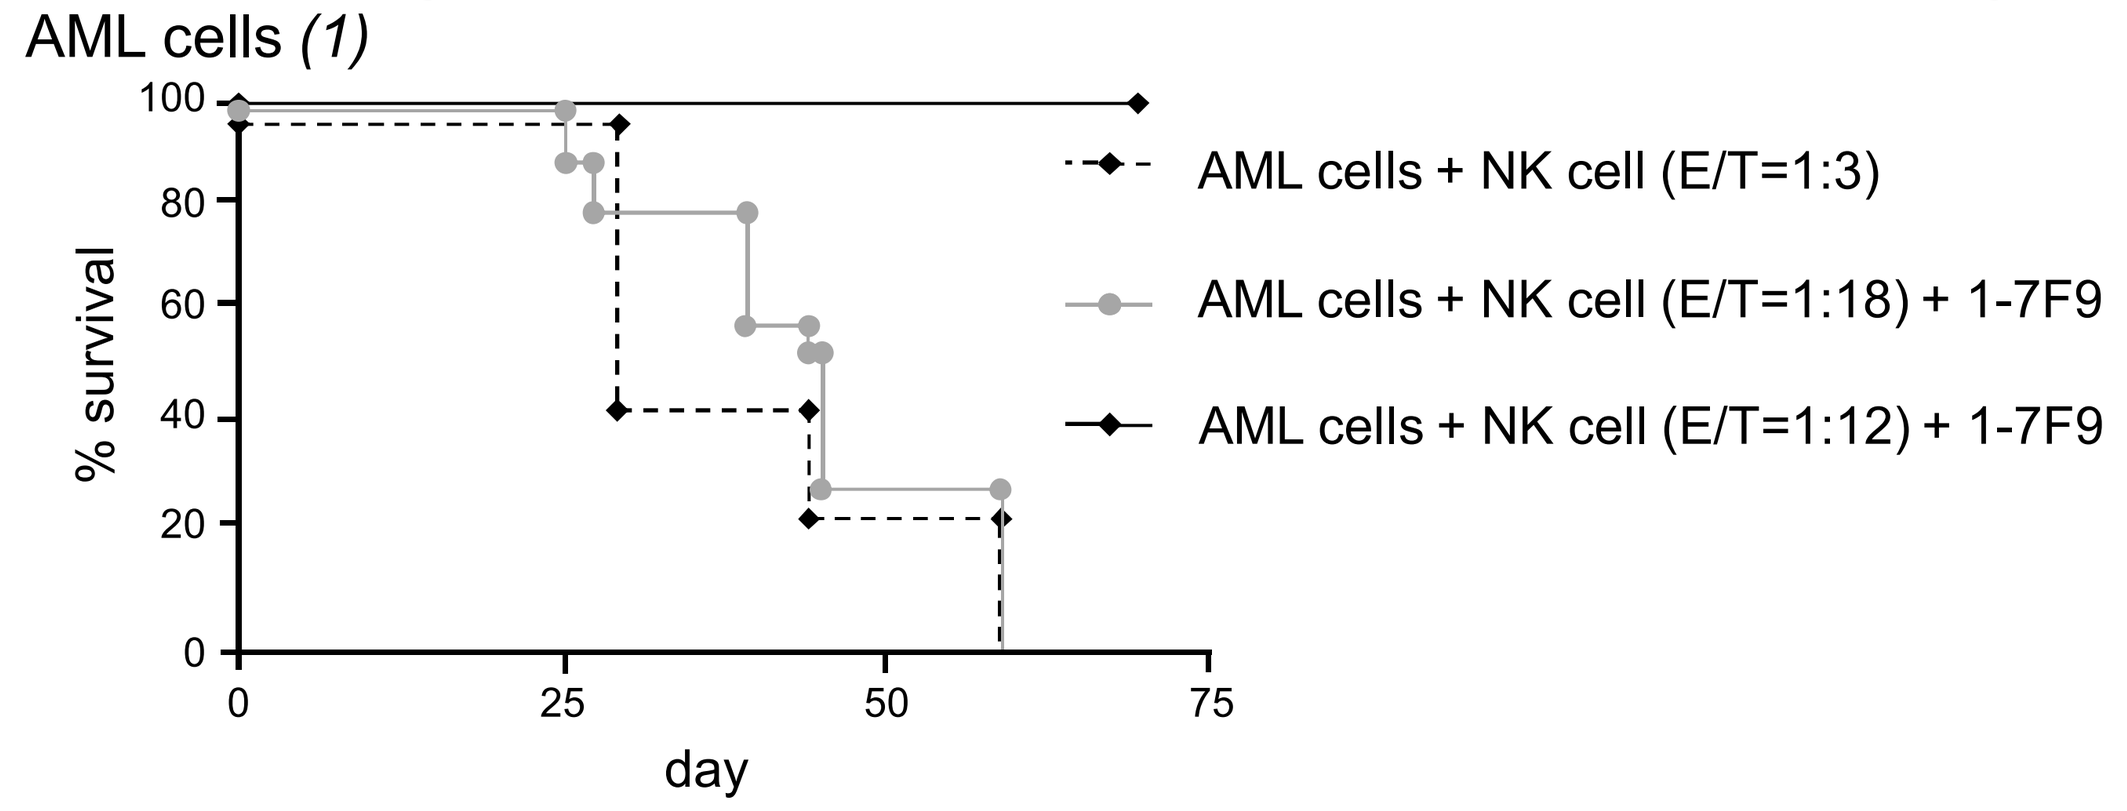
100
80
60
40
20
0
% survival
0
25
50
75
day
AML cells + NK cell (E/T=1:3)
AML cells + NK cell (E/T=1:18) + 1-7F9
AML cells + NK cell (E/T=1:12) + 1-7F9

Haploidentical Donors in Hematopoietic Cell Transplantation

- Lack of KIR-HLA class I interactions has been associated with potent NK-mediated antitumor efficacy and increased survival in patients with AML upon haplo-identical stem cell transplantation from KIR-mismatched donors
- AML patients transplanted with KIR-mismatched donor NK cells had lower relapse rates (3% versus 47%, p<0.01) and reduced risk of relapse with a relative risk of 0.48, 95% CI 0.29-0.78 *(2)*

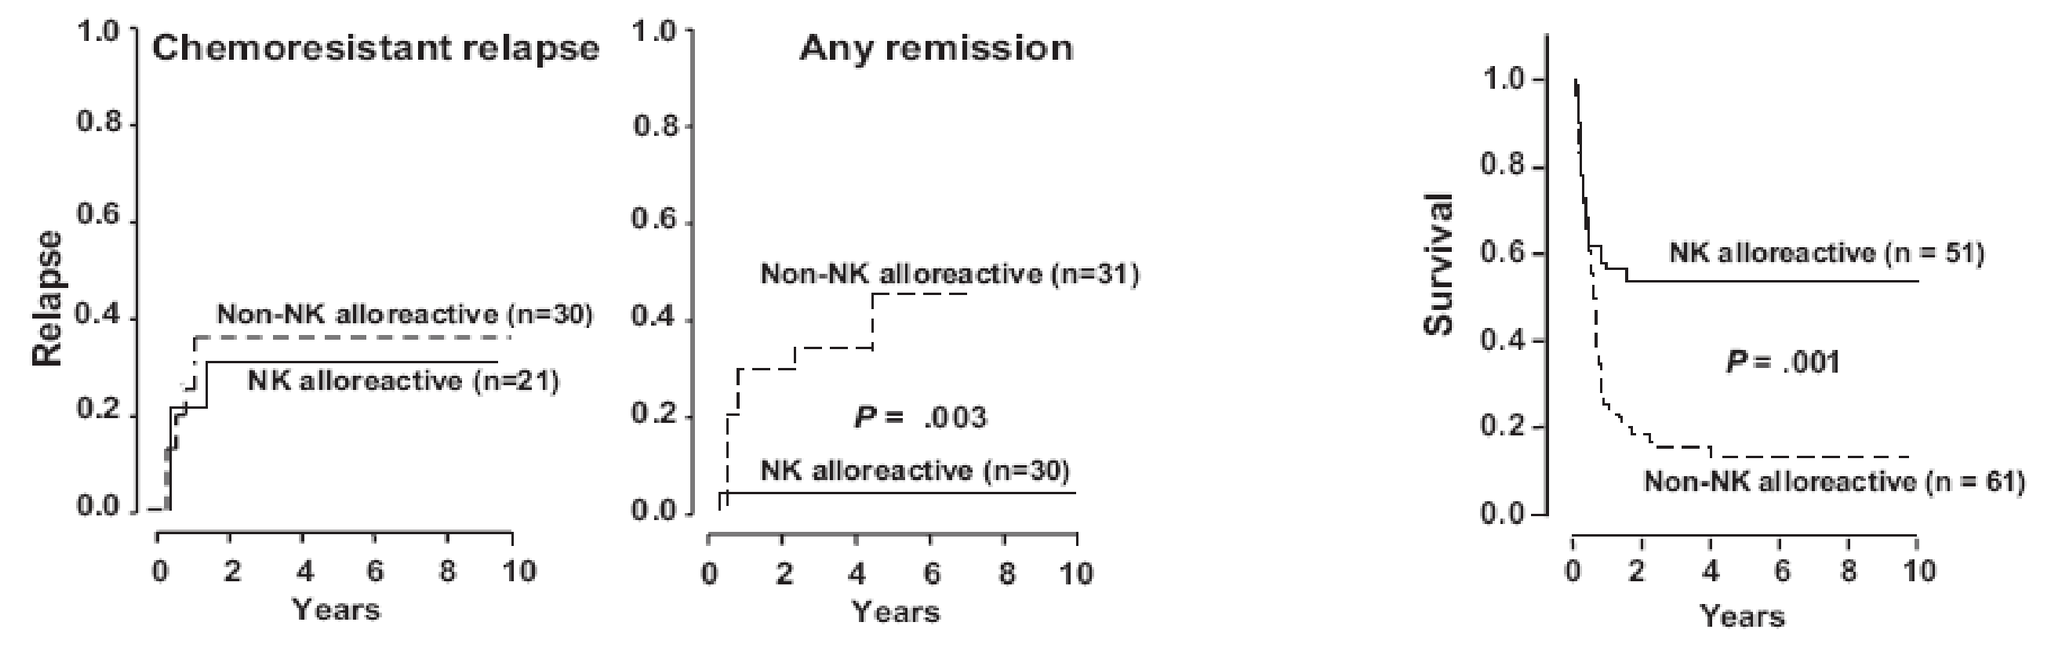
Chemoresistant relapse
Relapse
Non-NK alloreactive (n=30)
NK alloreactive (n=21)
Any remission
Non-NK alloreactive (n=31)
P = .003
NK alloreactive (n=30)
Survival
NK alloreactive (n = 51)
P = .001
Non-NK alloreactive (n = 61)
Years

Introduction

- A previously reported phase 1 trial conducted in elderly patients with AML in CR indicated a satisfactory safety profile *(3)*. The maximal tolerated dose was not reached at 3 mg/kg. There was a clear correlation between drug exposure and KIR occupancy. Survival was consistent with a dose-related anti-leukemia effect
- As experience with repeated cycles of IPH2101 was limited, the safety and efficacy of repeated monthly cycles of IPH2101 given at doses producing continuous KIR occupancy were further assessed in a cohort extension of the phase 1 trial
- The results of this extension study are reported here

Primary Objective of The Cohort Extension

- Further assessment of safety of repeated administrations of IPH2101 at doses expected to result in continuous KIR occupancy

Study Design

- Open- label, multicenter extension to a phase 1 dose-escalation study in elderly patients with acute myeloid leukemia in complete remission *(3)*
- 2 cohorts of 6 patients enrolled sequentially at 2 different doses of IPH2101
 - 1 and 2 mg/kg administered as 1-hour IV infusion
 - Repeated administrations every 4 weeks for a total of 6 administrations

Main Selection Criteria

- Age ≥ 60 years and ≤ 80 years
- AML in CR or CRi after 1 or 2 cycles of induction chemotherapy & 1-6 cycles of consolidation chemotherapy
- Exclusion of Acute Promyelocytic and Core Binding Factor Leukemia
- Ineligibility for allogeneic cell transplantation
- Time lapse ≤ 60 days since the last consolidation
- Recovery from toxicities of prior chemotherapies

Patient Characteristics

	1 mg/kg (n=6)	2 mg/kg (n=6)	Overall (n=12)
Median age in years (range)	71 (64-75)	71 (64-75)	71 (64-75)
Gender M:F	3:3	5:1	8:4
Median WBC at diagnosis (10^9/L) (range)	2.7 (1.2-31)	141 (1.6-322)	12.6 (1.2-322)
Intermediate cytogenetics (4)	6	6	12
FLT3 -ITD without NPM1 mutation	1	0	1
FLT3- ITD with NPM1 mutation	1	2	3
NPM1 mutation without FLT3-ITD	0	2	2
No identified molecular abnormality	4	1	5
Not done	0	1	1
Secondary AML (previous MDS)	0	1	1
Second CR	1	0	1
Median no. of consolidations (range)	3 (1-6)	2.5 (2-4)	3 (1-6)
Median Interval in weeks (range)			
Diagnosis - IPH2101	31 (17-113)	25 (18-29)	28 (17-113)
CR - IPH2101	24 (11-35)	20 (13-26)	21 (11-35)
Last consolidation - IPH2101	7 (5-10)	9 (7-13)	8 (5-13)
ECOG PS			
0	3	3	6
1	3	3	6

Treatment

- 7 patients received 6 cycles of IPH2101 and the 5 others between 1 and 3 cycles
- The first 6 patients received a dose of 1 mg/kg
- 6 additional patients received 2 mg/kg

	1 mg/kg (n=6)	2 mg/kg (n=6)	Overall (n=12)
No. of cycles			
1	0	1	1
2	0	1	1
3	3	0	3
6	3	4	7
Median no. of cycles (range)	4.5 (3-6)	6 (1-6)	6 (1-6)

KIR Occupancy by IPH2101

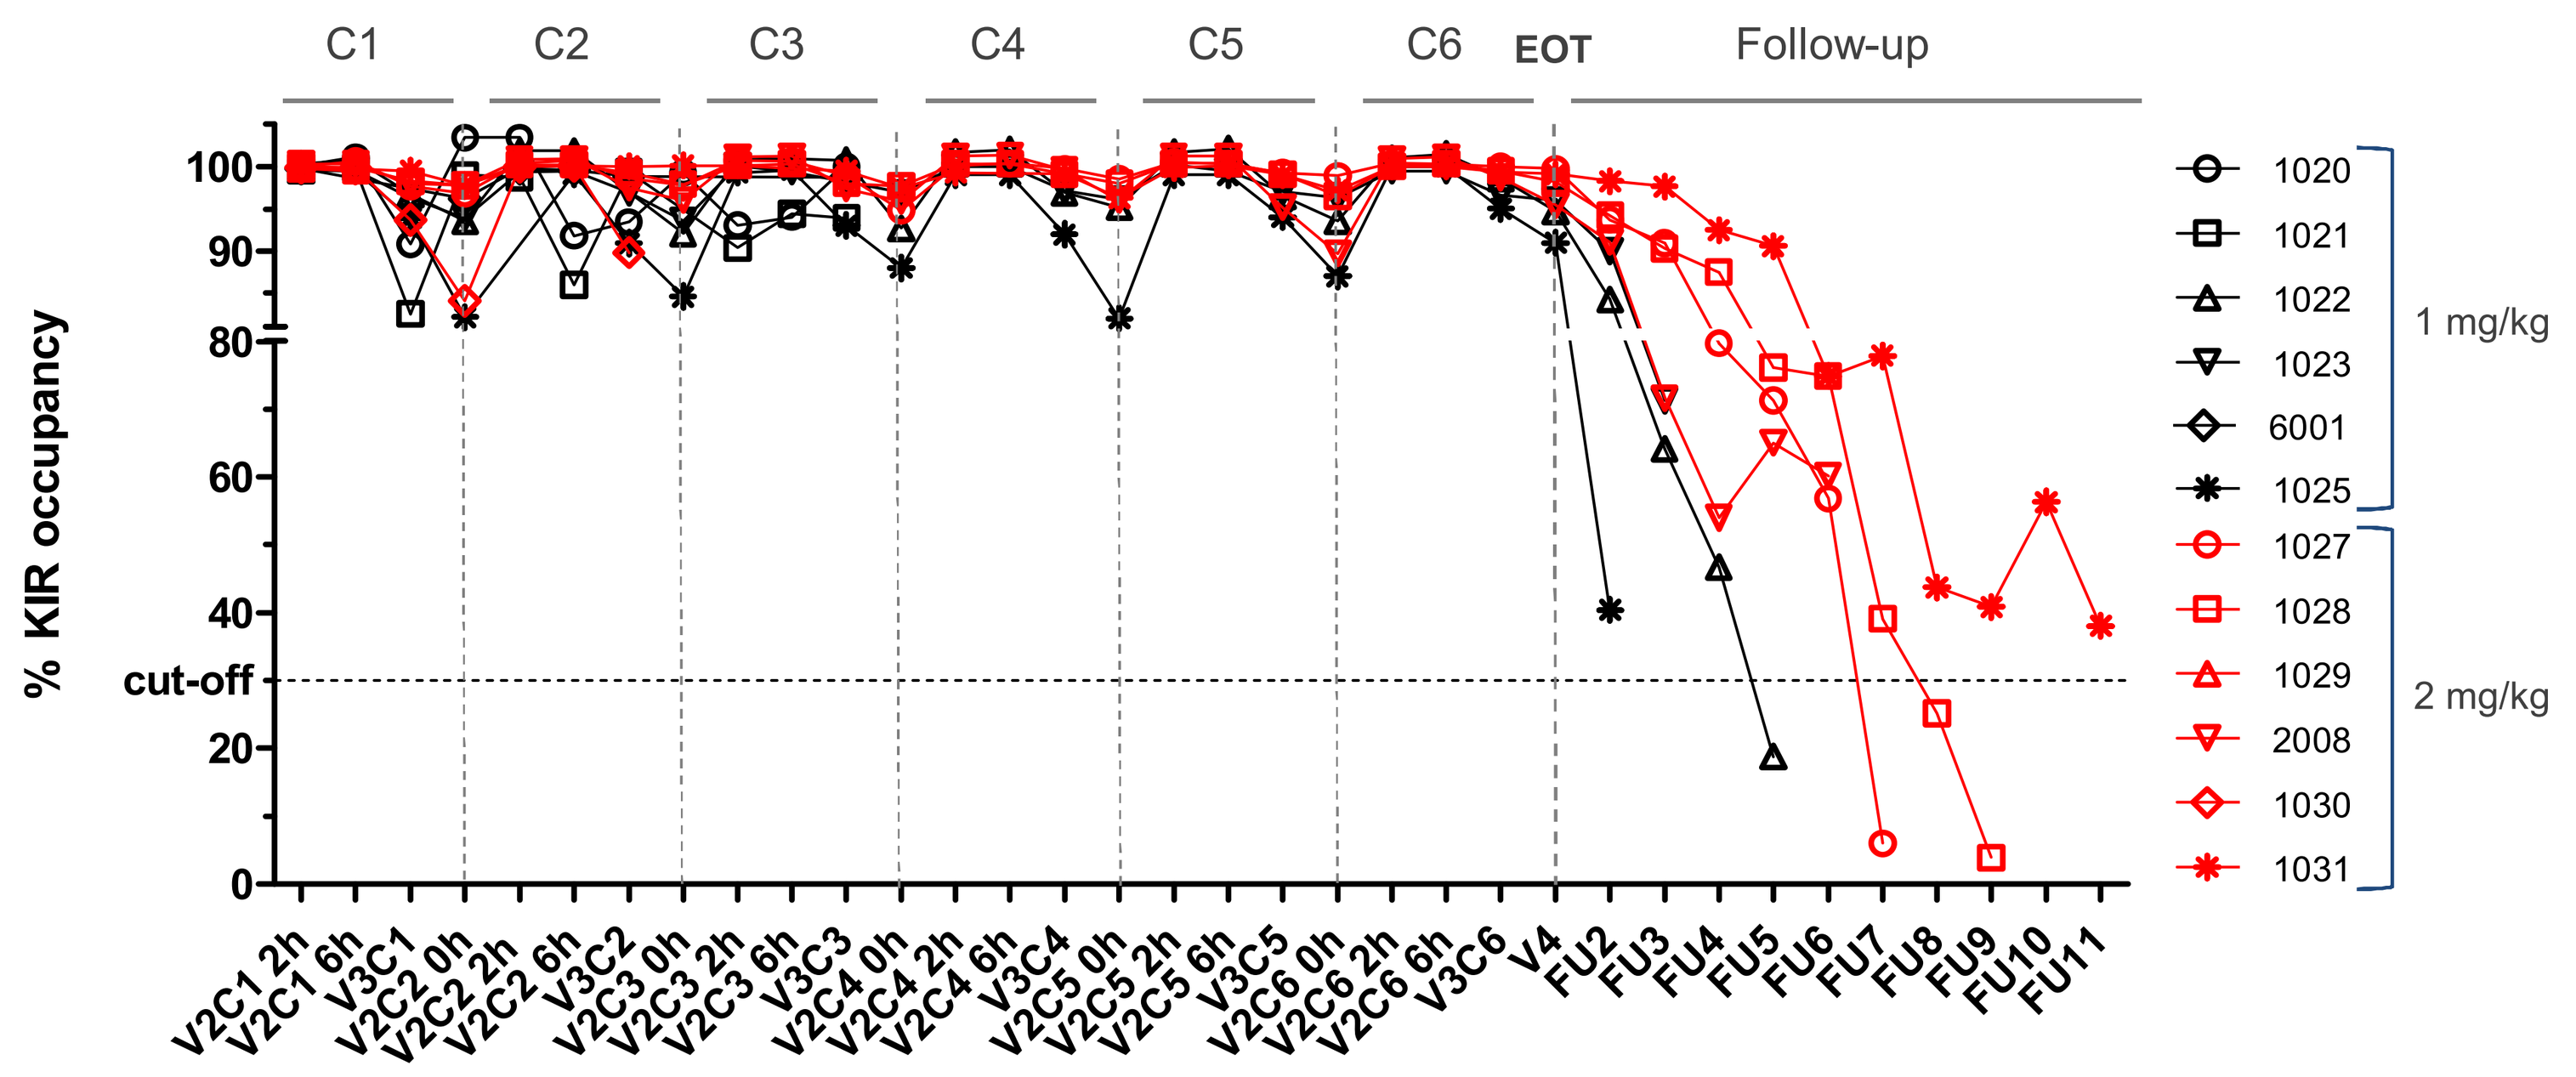
C1
C2
C3
C4
C5
C6
EOT
Follow-up
% KIR occupancy
100
90
80
60
40
cut-off
20
0
V2C1 2h
V2C1 6h
V3C1
V2C2 0h
V2C2 2h
V2C2 6h
V3C2
V2C3 0h
V2C3 2h
V2C3 6h
V3C3
V2C4 0h
V2C4 2h
V2C4 6h
V3C4
V2C5 0h
V2C5 2h
V2C5 6h
V3C5
V2C6 0h
V2C6 2h
V2C6 6h
V3C6
V4
FU2
FU3
FU4
FU5
FU6
FU7
FU8
FU9
FU10
FU11
1020
1021
1022
1023
6001
1025
1 mg/kg
1027
1028
1029
2008
1030
1031
2 mg/kg

Percentage KIR occupancy is the ratio of staining with PE-IPH2101 at the indicated time following treatment compared to the pre-dose staining.

Assays were performed by Platine PPS (Lyon, France)

Safety

- No death related to AEs
- Neither no related SAE nor SUSAR
- Grade 3 related AEs in 4 patients
 - Lymphopenia
 - Transient elevation of lipase and amylase
- No treatment discontinuation due to a related AE
 - 1 single treatment discontinuation due to unrelated cardiac insufficiency
- Limited number of recurrent grade 1-2 related AEs
 - Skin disorder (pruritus and/or rash/erythema) in 5 patients (10 events)

	1 mg/kg (n=6)	2 mg/kg (n=6)	Overall (n=12)
All AEs			
All grades	6 (100%)	6 (100%)	12 (100%)
Grade 4	2 (33%)	5 (83%)	7 (53%)
Grade 3	6 (100%)	5 (83%)	11 (92%)
Grade 2	5 (83%)	3 (50%)	8 (67%)
Grade 1	5 (83%)	6 (100%)	11 (92%)
Related AEs			
All grades	5 (83%)	5 (83%)	10 (83%)
Grade 4	0	0	0
Grade 3	1 (17%)	3 (50%)	4 (33%)
Grade 2	1 (17%)	1 (17%)	2 (17%)
Grade 1	4 (35%)	4 (35%)	8 (67%)

Related AEs CTCAE Preferred Term	Total	G1	G2	G3
Lymphopenia	4	0	0	4
Pruritus	3	3	0	0
Erythema	2	2	0	0
Lipase increase	1	0	0	1
Blood amylase increased	1	0	0	1
Flushing	1	0	1	0
Bundle branch block left	1	0	1	0
Rash	1	1	0	0
Pyrexia	1	1	0	0
Eye pruritus	1	1	0	0
Fatigue	1	1	0	0
Livedo reticularis	1	1	0	0
Back pain	1	1	0	0
Muscle spasms	1	1	0	0
Headache	1	1	0	0
Tremor	1	1	0	0
Diarrhea	1	1	0	0

Clinical Outcome

Dose	N*	PFS (months)	OS (months)
<1 mg/kg	16	2. 3	12.6
1-3 mg/kg	16	9.5	20.0
HR (95%CI)		0.515 (0.245; 1.081)	0.490 (0.219; 1.096)
P-value		0.075	0.076

*Combined data dose-escalation and cohort extension for patients in CR1

Includes updated data from the dose escalation phase 1 trial (1 patient in continuous CR for 85 months, and 3 patients still alive after 58, 60 and 63 months, respectively)

In the cohort extension, 2 patients are still in continuous CR after 18 and 21 months, respectively, and 1 patient is still alive after 42 months; 1/12 patient was excluded from this analysis (relapse within 5 days).

Conclusions

- Satisfactory safety of IPH2101 confirmed in this extension trial, while monthly cycles led to full and sustained KIR occupancy > 6 months
- The most frequent AEs consisted of mild to moderate reactions related to the infusion (no pre-medication)
- There were no immune-related disorders, apart from mild to moderate transient skin disorders
- KIR occupancy: moderate inter-patient and inter-cycle variability with maximal occupancy (100%) reached in 2 hours post-administration with both doses
- Trends in PFS and OS at <1mg/kg vs ≥1 mg/kg are consistent with a dose effect of IPH2101
- An ongoing randomized phase 2 trial (EFFIKIR NCT 01687387) will assess prospectively the impact of anti-KIR therapy on clinical outcome in elderly patients with AML in CR1

References

1. Romagné F et al. *Blood* 2009; 114: 2667-74;
2. Ruggeri L et al. *Blood* 2007;110:433-40;
3. Vey N et al. *Blood* 2012; 120: 4317-4323;
4. Döhner H et al. *Blood* 2010; 115: 453-74.

RECEIVE A PDF OF THIS POSTER ON YOUR MOBILE PHONE

Copies of this poster obtained through Quick Response Code are for personal use only and may not be reproduced without permission from ASH and the author of this poster

ASH – December 2013

Disclosures: Vey: *Innate Pharma:* Honoraria; *Bristol Myers Squibb:* Honoraria. Andre: *Innate-Pharma:* Employment, Equity Ownership. Calmels: *Innate-Pharma:* Employment, Equity Ownership. Zerbib: *Innate Pharma:* Employment. Buffet: *Innate-Pharma:* Employment.

innate pharma