Anti-KIR antibody enhancement of anti-lymphoma activity of natural killer cells as monotherapy and in combination with anti-CD20 antibodies

LEUKEMIA & LYMPHOMA SOCIETY®
fighting blood cancers

Abstract # 4417

Holbrook E. Kohrt [1*], Ariane Thielens [2*], Aurelien Marabelle [1], Idit Sagiv-Barfi [1], Caroline Sola [2], Fabien Chanuc [2], Nicolas Fuseri [2], Cécile Bonnafous [2], Debra Czerwinski [1], Amanda Rajapaksa [1], Erin Waller [1], Sophie Ugolini [3], Eric Vivier [3], François Romagné [2], Ronald Levy [1*], Pascale André [2*] and Mathieu Bléry [2*]

1- Department of Medicine, Division of Oncology, Stanford University, Stanford, CA **2-** Innate Pharma, Marseille, France **3 -** Centre d'Immunologie de Marseille-Luminy, INSERM U1104, CNRS UMR7280, Aix Marseille Université UM2, Marseille, France ***authors contributed equally to work**

Abstract

- Natural killer (NK) cells mediate anti-lymphoma activity by spontaneous cytotoxicity and antibody-dependent cell-mediated cytotoxicity (ADCC) when triggered by rituximab, an anti-CD20 monoclonal antibody (mAb) used to treat patients with B cell lymphomas. The balance of inhibitory and activating signals determines the magnitude of NK cell's efficacy by spontaneous cytotoxicity.
- Here, using a killer cell immunoglobulin-like receptor (KIR) transgenic murine model, we show that blockade of the interface of inhibitory KIRs with MHC class I antigens on lymphoma by anti-KIR antibodies prevents a tolerogenic interaction and augments NK cell spontaneous cytotoxicity.
- In combination with anti-CD20 mAbs, anti-KIR treatment induces enhanced NK cell-mediated, rituximab-dependent cytotoxicity against lymphoma *in vitro* and *in vivo* in KIR transgenic and syngeneic murine lymphoma models.
- These results support a therapeutic strategy of combination, rituximab and KIR blockade through lirilumab, illustrating the potential efficacy of combining a tumor targeting therapy with an NK cell agonist thus stimulating the post-rituximab anti-lymphoma immune response.

Mechanism of action

Anti-KIR mAb blocks an inhibitory signal to NK cells and enhances rituximab dependent cytotoxicity (ADCC)

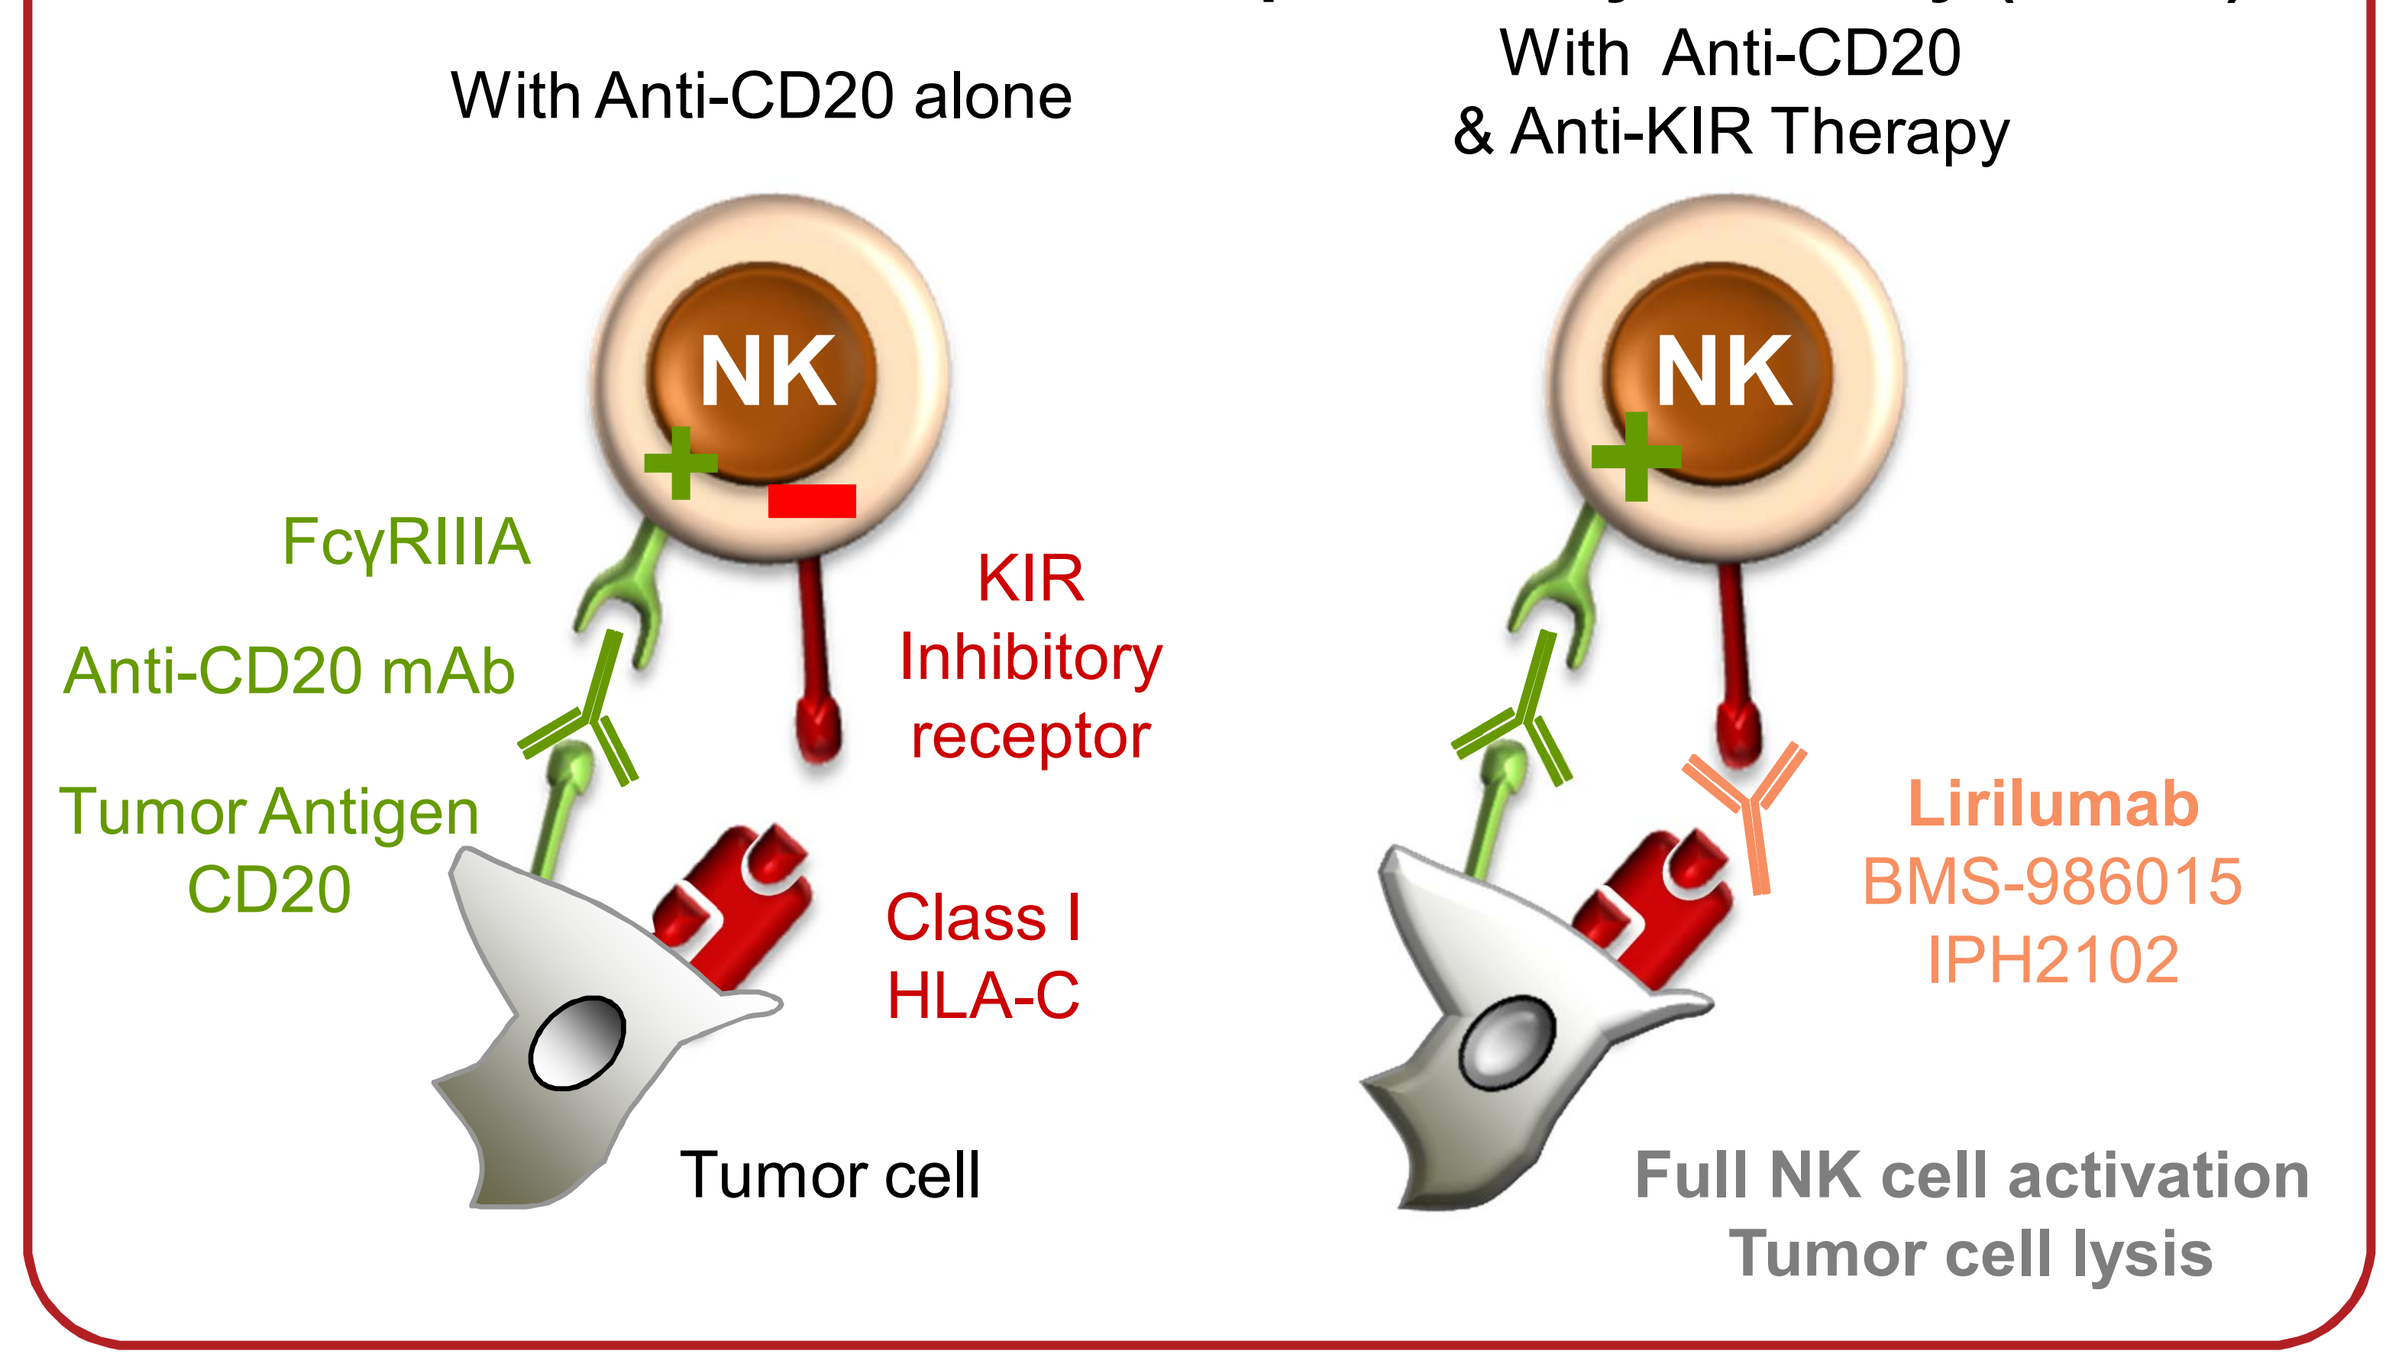

With Anti-CD20 alone
With Anti-CD20 & Anti-KIR Therapy
NK
NK
FcγRIIIA
Anti-CD20 mAb
Tumor Antigen CD20
KIR Inhibitory receptor
Class I HLA-C
Tumor cell
Lirilumab
BMS-986015
IPH2102
Full NK cell activation
Tumor cell lysis

Results

Anti-Ly49C/I F(ab')$_2$ increases anti-CD20 mAb-mediated NK cell degranulation and cytotoxicity

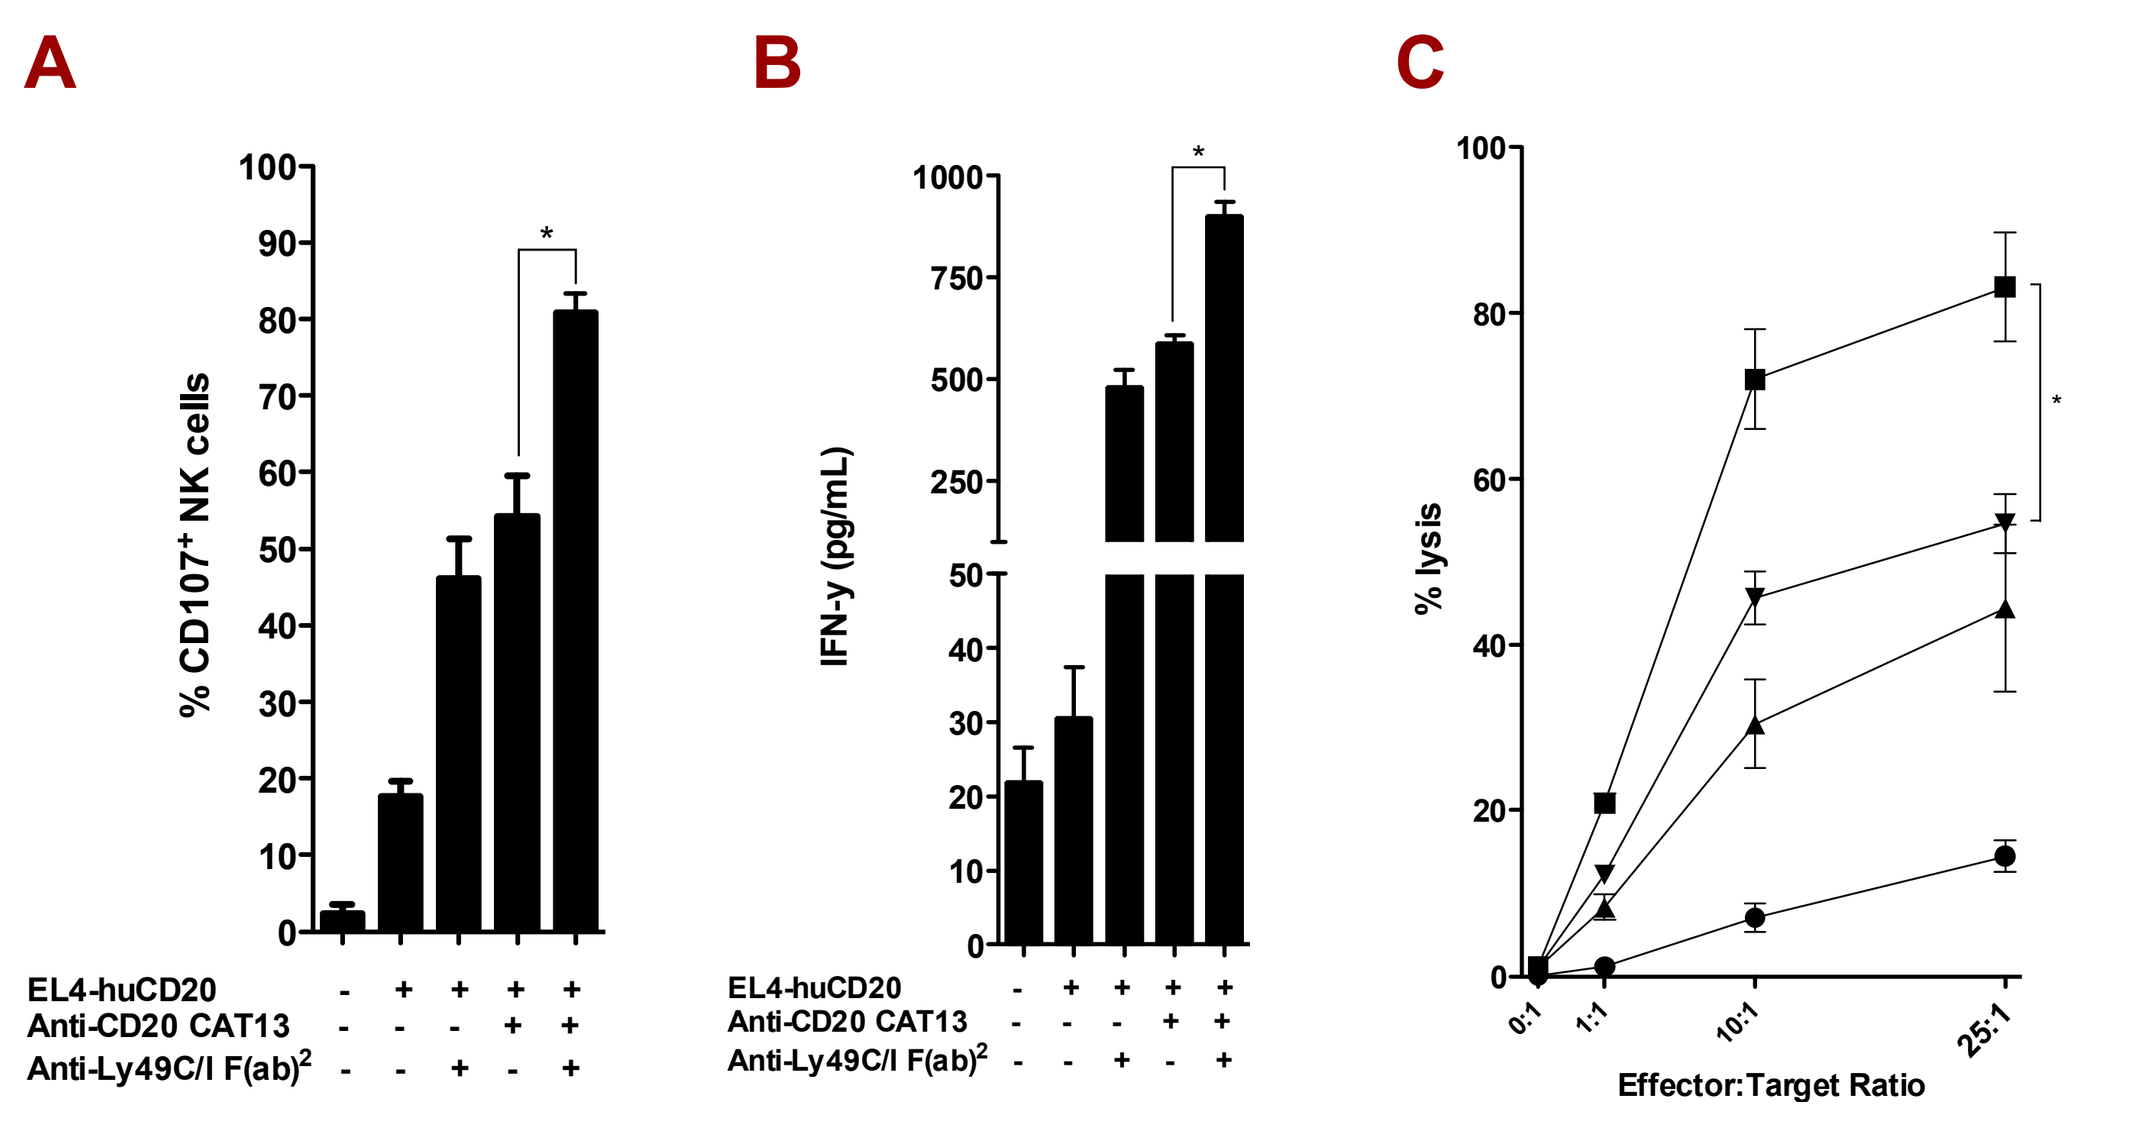

A
B
C
% CD107+ NK cells
IFN-γ (pg/mL)
% lysis
EL4-huCD20
Anti-CD20 CAT13
Anti-Ly49C/I F(ab)2
Effector:Target Ratio

- Purified NK cells from C57BL/6 mice were analyzed: (**A**) for degranulation by **CD107a mobilization** after 24 hour culture with five conditions: medium alone, human CD20-positive murine lymphoma cell line (EL4-huCD20) at a 1:1 ratio of NK cells: tumor cells, tumor and anti-CD20 mAb (CAT13, 10 µg/mL), tumor and anti-Ly49C/I F(ab')$_2$ (5E6, 10 µg/mL), or tumor, anti-CD20 mAb (10 µg/mL), and anti-Ly49C/I F(ab')$_2$ (10 µg/mL) (A, **p*=.0109), (**B**) **for IFN-γ production** (ELISA) following a 4 hour culture in the conditions described above (B, **p*=.001), and (**C**) in **cytotoxicity assay** with chromium labeled EL4-huCD20; percent lysis of target cells at varying E:T cell ratios cultured with medium alone (●), anti-Ly49C/I F(ab')$_2$ (10 µg/mL, ▲), anti-CD20 mAb (10 µg/mL, ▼), or the combination of both (■) antibodies (C, **p*=.019).

Anti-Ly49C/I F(ab')$_2$ enhances the anti-lymphoma activity of anti-CD20 mAb *in vivo* and is NK cell-dependent

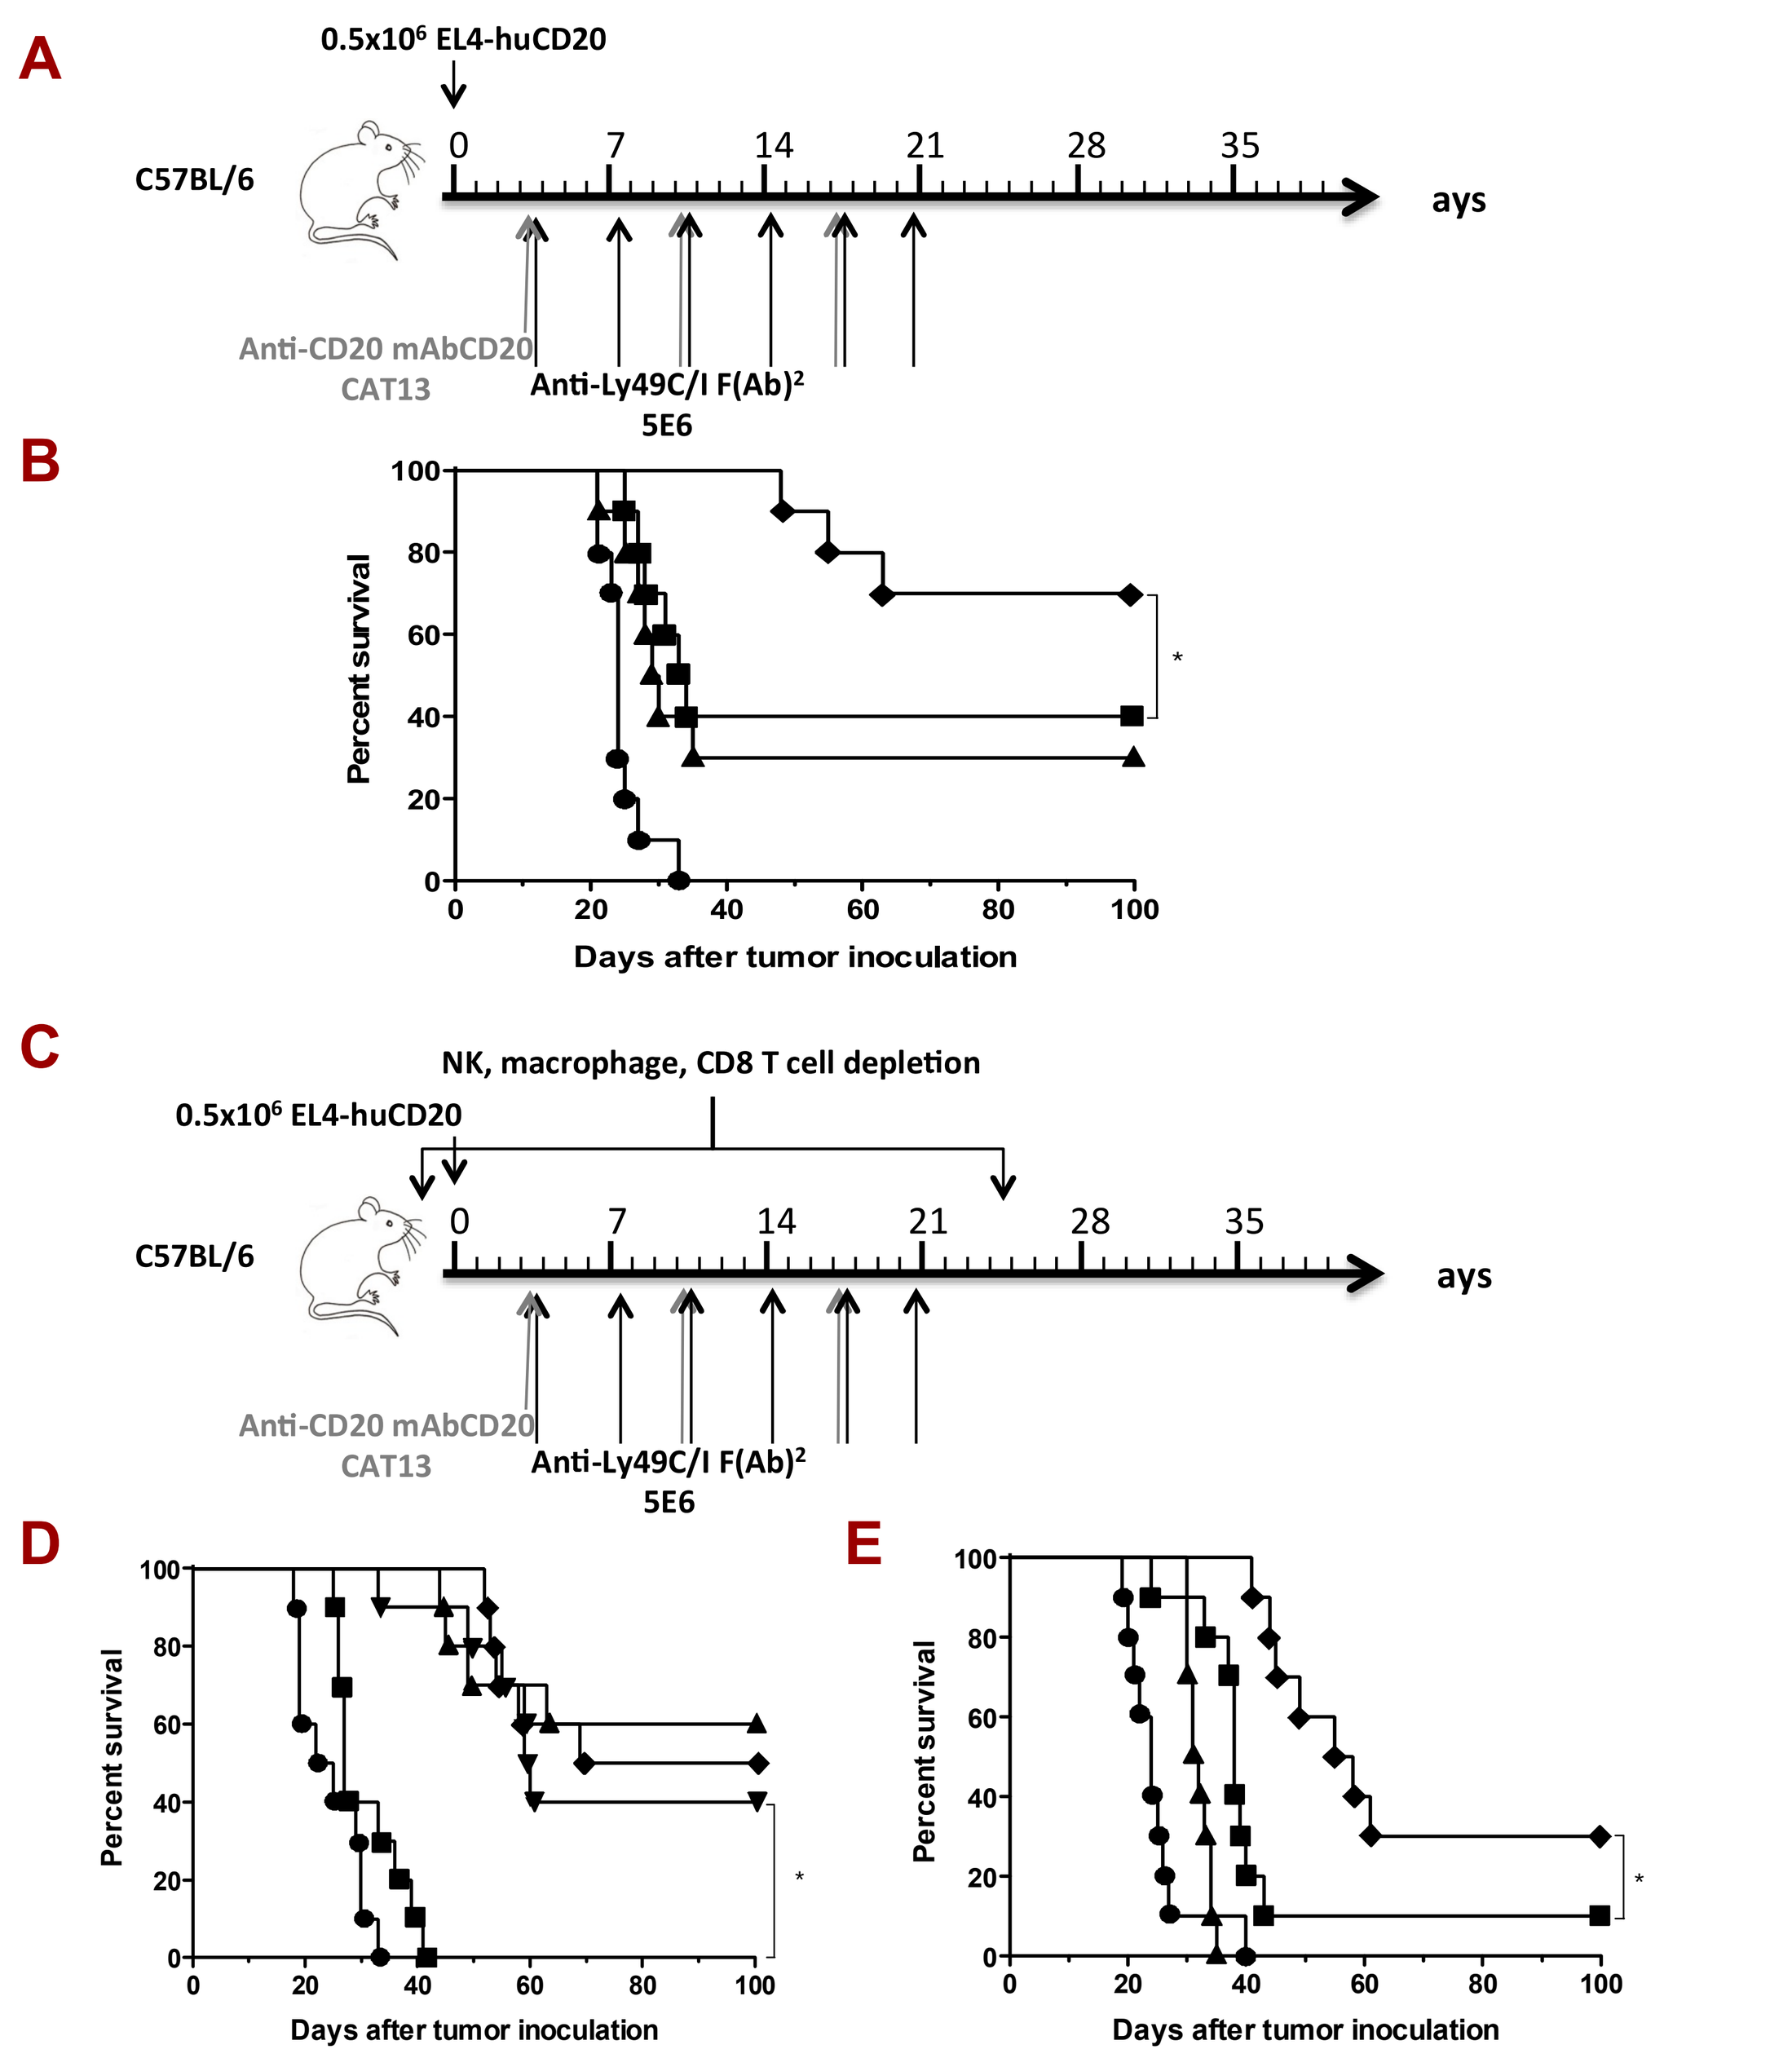

A
0.5x10⁶ EL4-huCD20
C57BL/6
Anti-CD20 mAbCD20 CAT13
Anti-Ly49C/I F(Ab)2 5E6
B
Percent survival
Days after tumor inoculation
C
NK, macrophage, CD8 T cell depletion
D
E

- (**A-B**) Post-tumor inoculation, mice received either Rat IgG control (●), 250 µg of anti-CD20 mAb (CAT-13) on day 3 and weekly for three weeks (■), 200 µg of anti-Ly49C/I F(ab')$_2$ (5E6) on day 3 (▲) and biweekly for three weeks, or the combination (◆). Mice were then monitored for overall survival (B, **p*=.042).
- (**C-D**) Post-tumor inoculation, mice received either Rat IgG control on day 3 (●), 50µL of anti-Asialo-GM1 on days -1, 0, 5, 10,15, 20, and 25 with 250 µg of anti-CD20 mAb and 200 µg anti-Ly49C/I F(ab')$_2$ on day 3 (■), 200µL liposomal (L.) clodronate on day -2 followed by 100µL on days 0, 4, 8,12, 16, 20, and 24 with 250 µg of anti-CD20 mAb and 200 µg of anti-Ly49C/I F(ab')$_2$ on day 3 (▼), 500 µg anti-CD8 mAb on days -1, 0, 5, 10,15, 20, and 25 with 250 µg of anti-CD20 mAb and 200 µg anti-Ly49C/I F(ab')$_2$ on day 3 (♦), or 250 µg of anti-CD20 mAb on day 3 and 200µg of anti-Ly49C/I F(ab')$_2$ on day 3 (▲) with 250µg of anti-CD20 mAb repeated weekly for three weeks and 200 µg of anti-Ly49C/I F(ab')$_2$ repeated biweekly for three weeks. Mice were then monitored for overall survival (D, **p*<.001, n=10 mice per group).
- (**E**) shows survival with identical treatment groups (A-B), however with treatment delayed until day 10 post-tumor inoculation (E, **p*=.005).

Lirilumab therapy improves survival in a therapeutic HLA⁺ tumor model in an NK cell-dependent manner

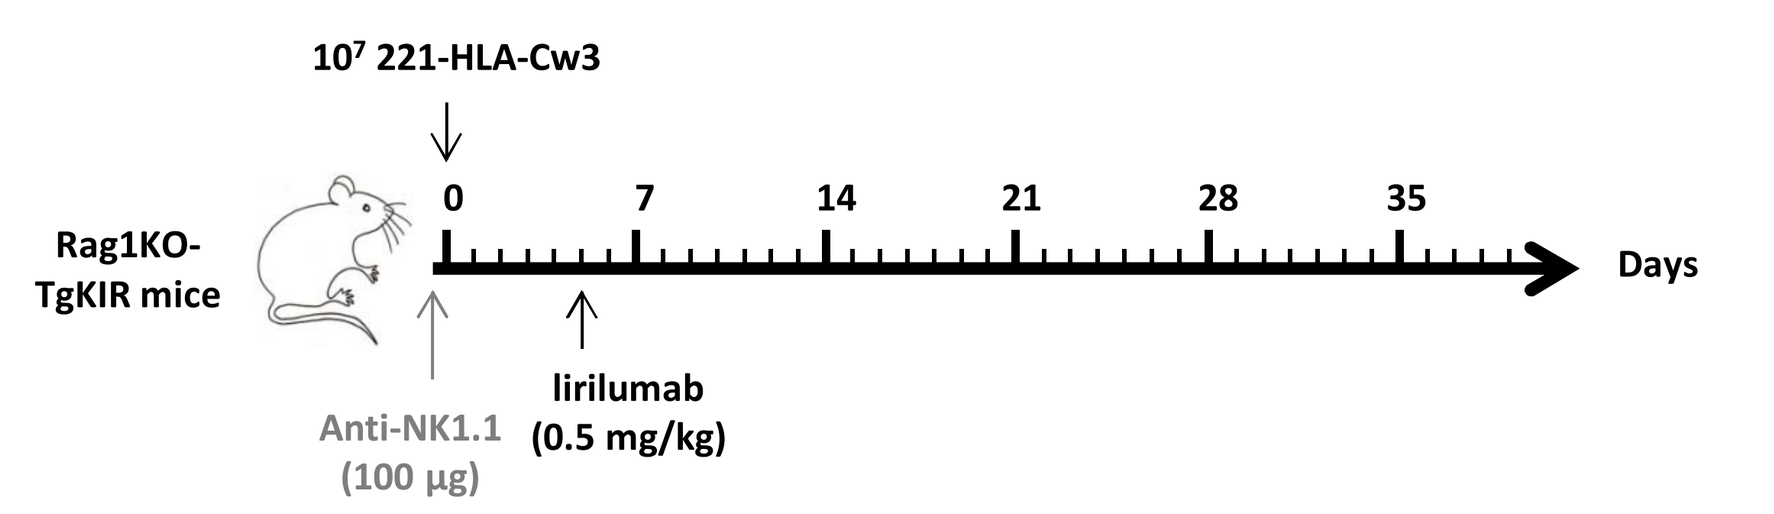

10⁷ 221-HLA-Cw3
Rag1KO-TgKIR mice
Days
Anti-NK1.1 (100 µg)
lirilumab (0.5 mg/kg)

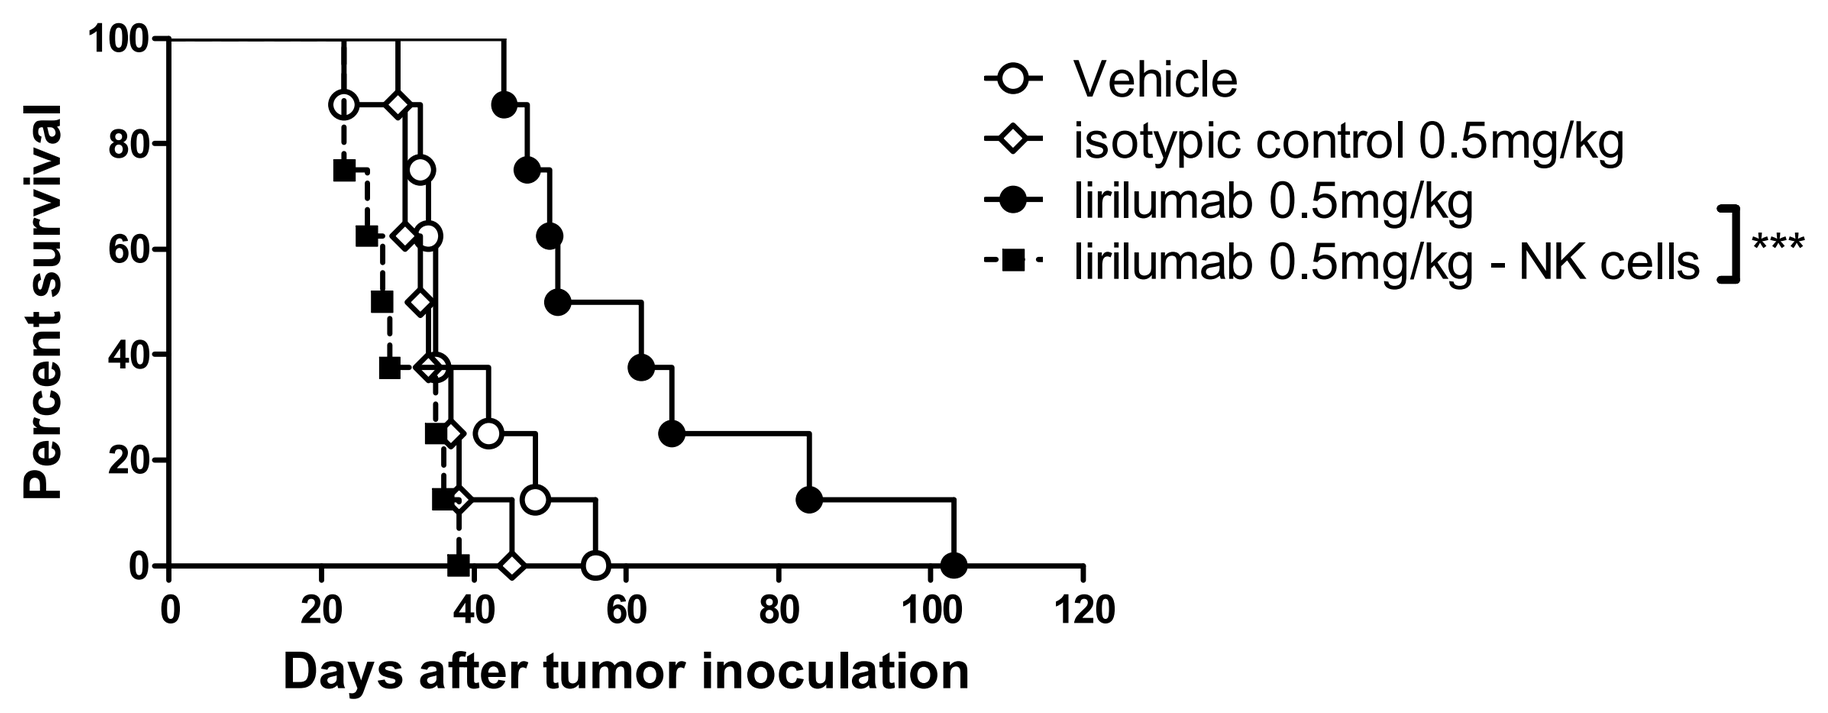

Percent survival
Days after tumor inoculation
Vehicle
isotypic control 0.5mg/kg
lirilumab 0.5mg/kg
lirilumab 0.5mg/kg - NK cells

- Rag1KO-Tg KIR mice (n=8 mice/group) received, starting five days after tumor challenge, either no treatment (o), isotype control at 0.5 mg/kg (◊), lirilumab at 0.5 mg/kg (●), or lirilumab at 0.5 mg/kg and 100 µg anti-NK1.1 mAb (■) to deplete NK cells starting on day prior to tumor inoculation. Mice were then monitored for overall survival (**p*=.0002).

Lirilumab therapy enhances the anti-lymphoma activity of rituximab *in vivo* and is NK cell-dependent

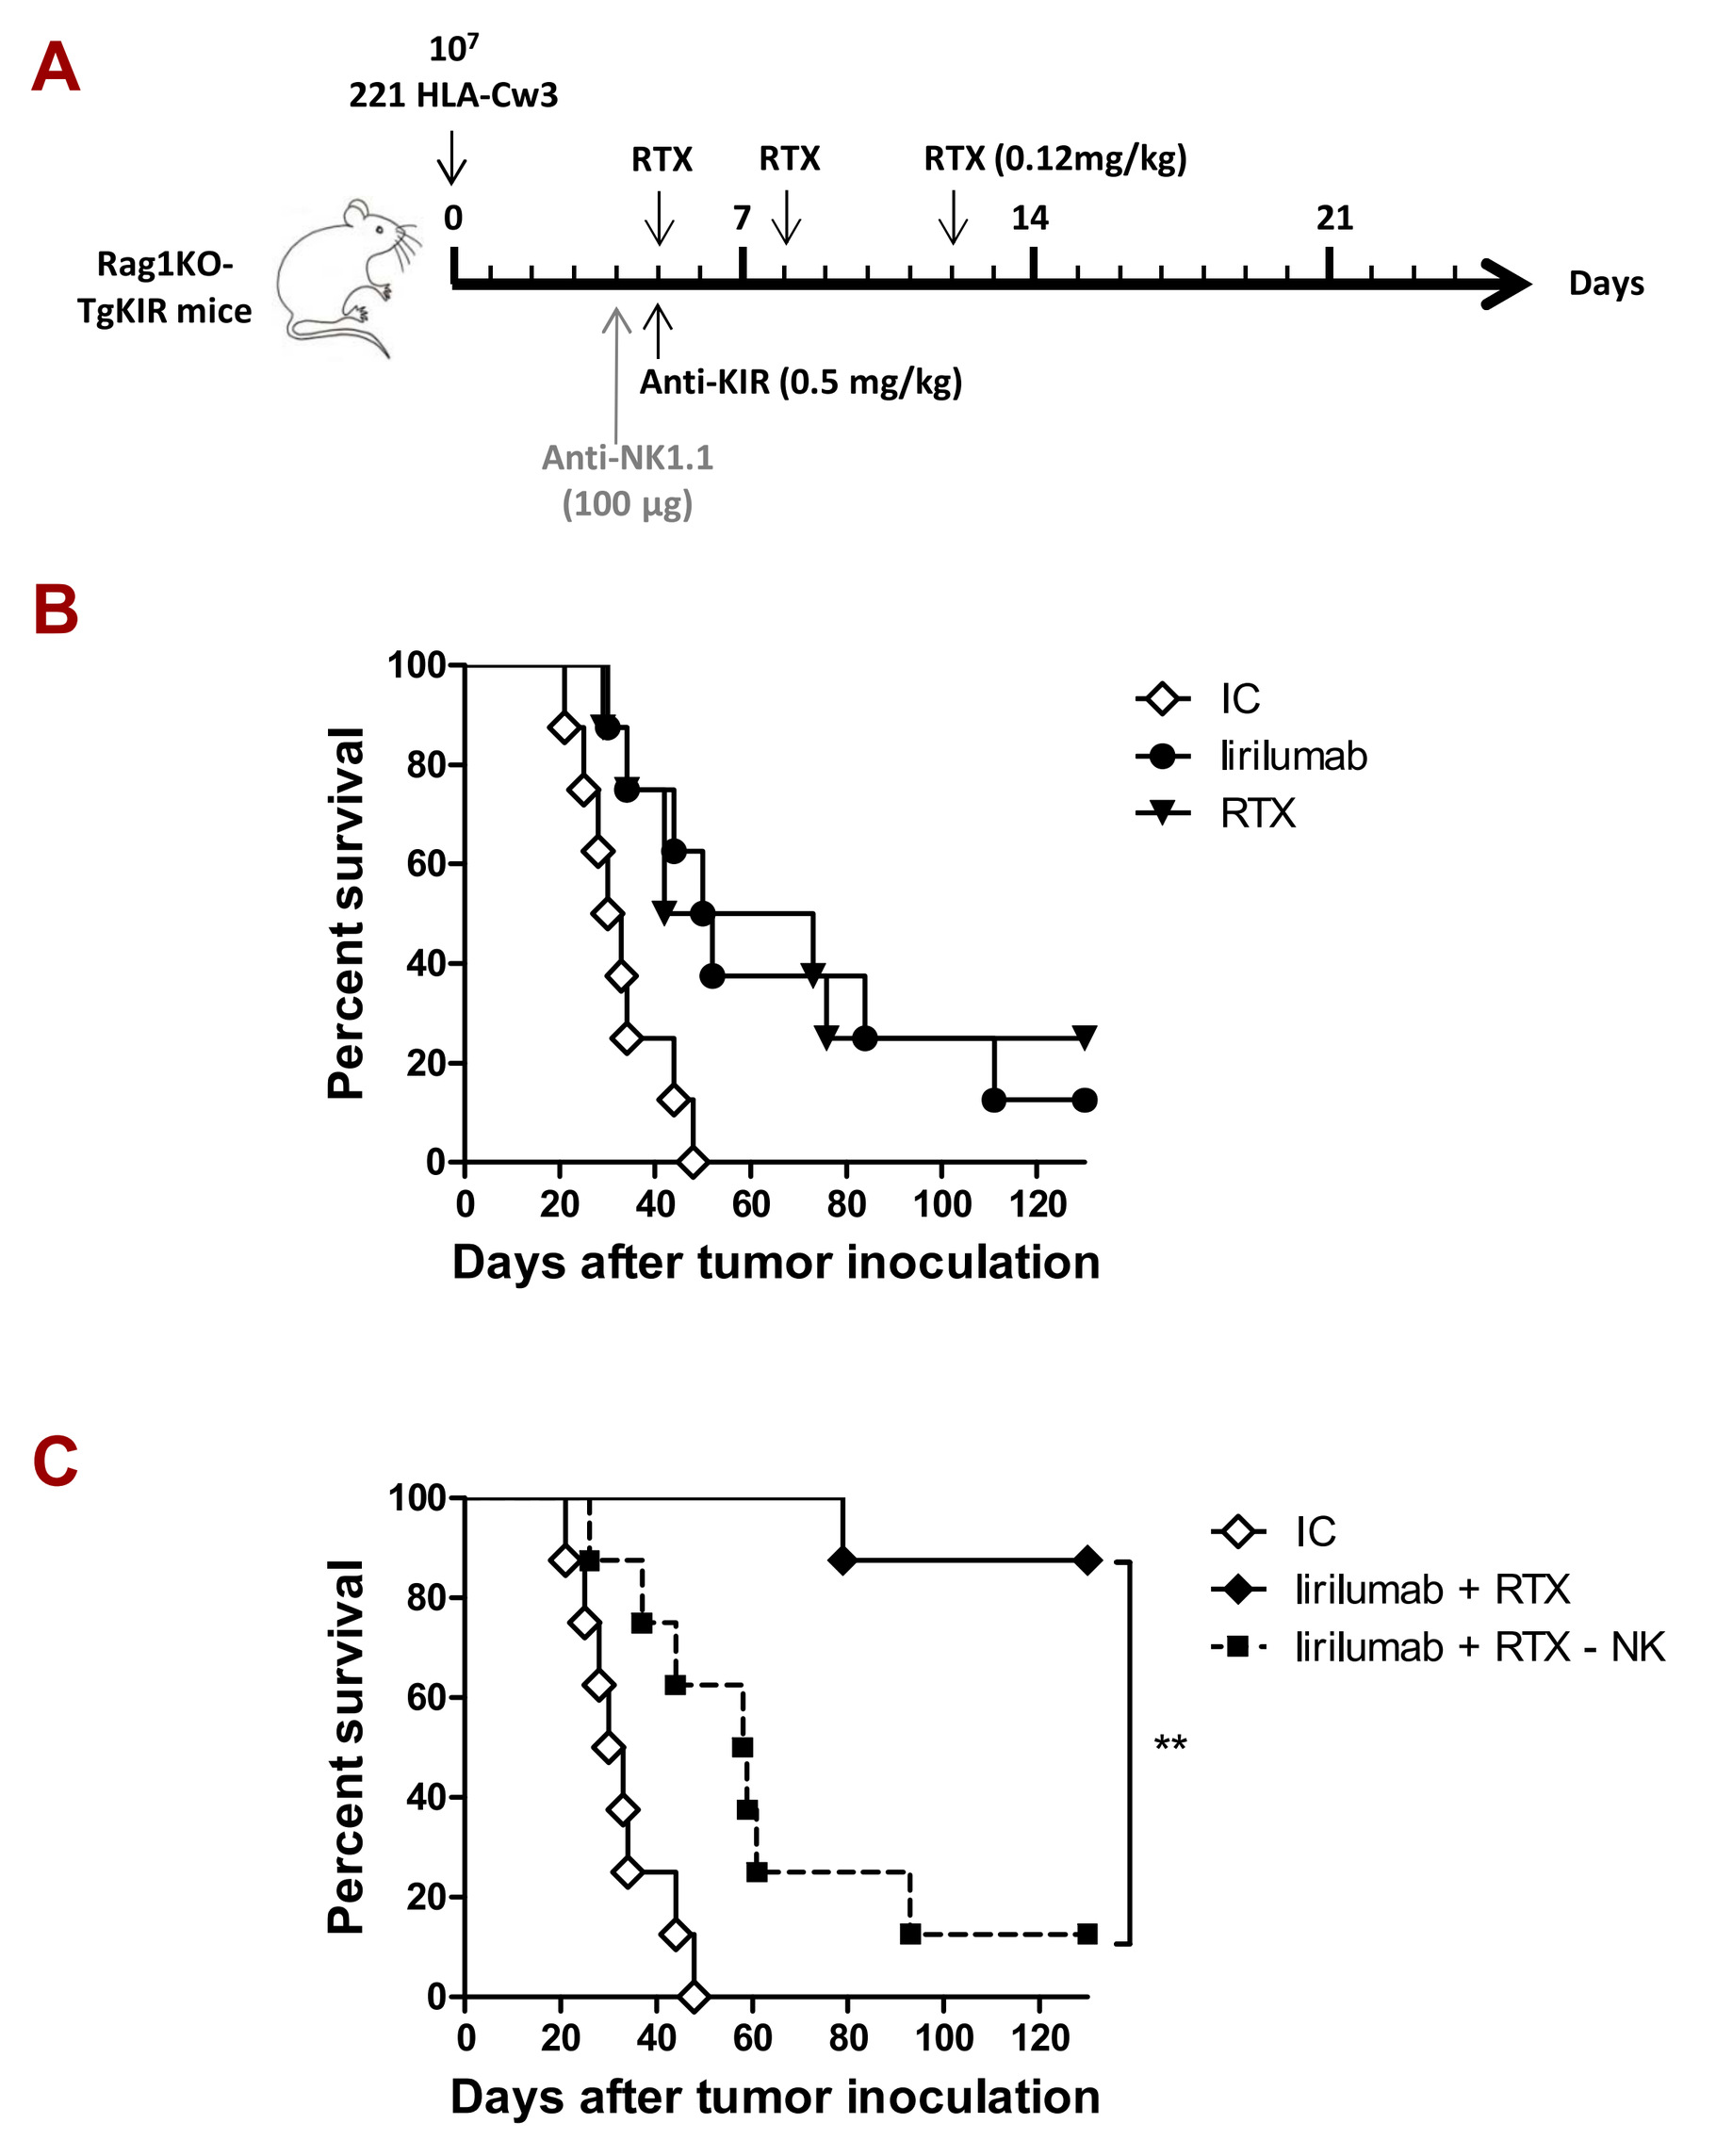

A
10⁷ 221 HLA-Cw3
RTX
RTX
RTX (0.12mg/kg)
Rag1KO-TgKIR mice
Days
Anti-KIR (0.5 mg/kg)
Anti-NK1.1 (100 µg)
B
IC
lirilumab
RTX
Percent survival
Days after tumor inoculation
C
IC
lirilumab + RTX
lirilumab + RTX - NK
**

- (**A-B**) Starting five days after tumor challenge, Rag1KO-Tg KIR mice (n=8 mice/group) received either isotype controls (◊), lirilumab i.v. at 0.5 mg/kg on day 5 (●), or rituximab i.p. at 0.12 mg/kg on days 5, 8 and 12 (▼) Mice were then monitored for overall survival.
- (**C**) To determine the role of NK cells, Rag1KO-Tg KIR mice (n=8 mice/group) received either isotype controls (◊), lirilumab at 0.5 mg/kg on day 5 and rituximab at 0.12 mg/kg on days 5, 8 and 12 (♦), or lirilumab at 0.5 mg/kg on day 5 and rituximab at 0.12 mg/kg on days 5, 8 and 12 and 100 µg of anti-NK1.1 mAb on day 4 (■) to deplete NK cells Mice were then monitored for overall survival (C, **p*<.001).

Conclusions

- We present here pre-clinical evidence supporting another novel approach to enhancing mAb antitumor efficacy.
- Our findings provide rationale for the combination of an anti-KIR mAb to "remove the brakes" from NK cells and further enhance the efficacy of rituximab among other mAbs by augmentation of ADCC.
- A clinical trial investigating this strategy is needed.

References

- Borgerdin A. et al., *Exp Hematol* 2011 38: 213-21
- Binyamin L. et al., 2008 *J Immunol* 180: 6392-401
- Romagné F. et al., *Blood* 2009; 114: 2667-74
- Kohrt H. et al., *Blood* 2011; 117: 2423-32

RECEIVE A PDF OF THIS POSTER ON YOUR MOBILE PHONE

Copies of this poster obtained through Quick Response Code are for personal use only and may not be reproduced without permission from ASH and the author of this poster

ASH – December 2013

Disclosures: A.T., C.S., F.C., N.F., C.B., F.R., P.A. and M.B. are employees of Innate Pharma. E.V. and F.R. are co-founders of Innate Pharma. The remaining authors declare no competing financial interests.

innate pharma