A phase 1 dose-escalation study of lirilumab (IPH2102, BMS-986015, LIRI), a fully human anti-KIR monoclonal antibody in patients with various hematologic (HEM) or solid (SOL) malignancies

Abstract ID #: 3065
Board #: 391

Copies of this poster obtained through Quick Response (QR) Code are for personal use only and may not be reproduced without permission from ASCO® and the author of this poster

N. Vey[1], L. Karlin[2], A. Gonçalves[1], S. Sadot-Lebouvier[3], F. Broussais[3], D. Marie[4], D. Berton-Rigaud[3], P. André[4], R. Zerbib[4], R. Buffet[4], T. Prebet[1], A. Charbonnier[1], J. Rey[1], A. Pigneux[5], J. Bennouna[3], N. Boissel[6] and G. Salles[2]
[1] Institut Paoli-Calmettes, Marseille, France; [2] C.H.U. de LYON Sud, Pierre Bénite, France; [3] ICO – Site René Gauducheau , St Herblain, France; [4] Innate Pharma, Marseille, France; [5] Centre Francois Magendie, H. Haut-Lévêque, Pessac, France; [6] H. St Louis, CIC, Paris, France

Background

Lirilumab (LIRI) is a fully human IgG4 monoclonal antibody (CHO manufacturing) which binds specifically and with high affinity to the main human inhibitory killer cell immunoglobulin-like receptors (KIR), KIR2DL1 and KIR2DL2/3, expressed by NK cells. This binding blocks the interaction of the KIR2DL receptors with HLA-C allotypes, their natural ligands, and prevents the inhibitory signals usually triggered by this interaction. The blockade of KIR by LIRI fosters the activation of NK cells, selectively enhancing the cytotoxicity of NK cells against tumor cells without affecting healthy tissues.

Experimental[1] and clinical[2] data suggest that enhancement of NK cell activity, notably by a mismatch between donor KIR and patient KIR ligand HLA-C, i.e. by a surrogate of KIR blockade, is susceptible to improve the prognosis of cancers after a reduction of tumor burden by previously administered treatments. We previously reported the safety and potential clinical activity of anti-KIR monoclonal antibody using IPH2101[3,4] (hybridoma manufacturing).

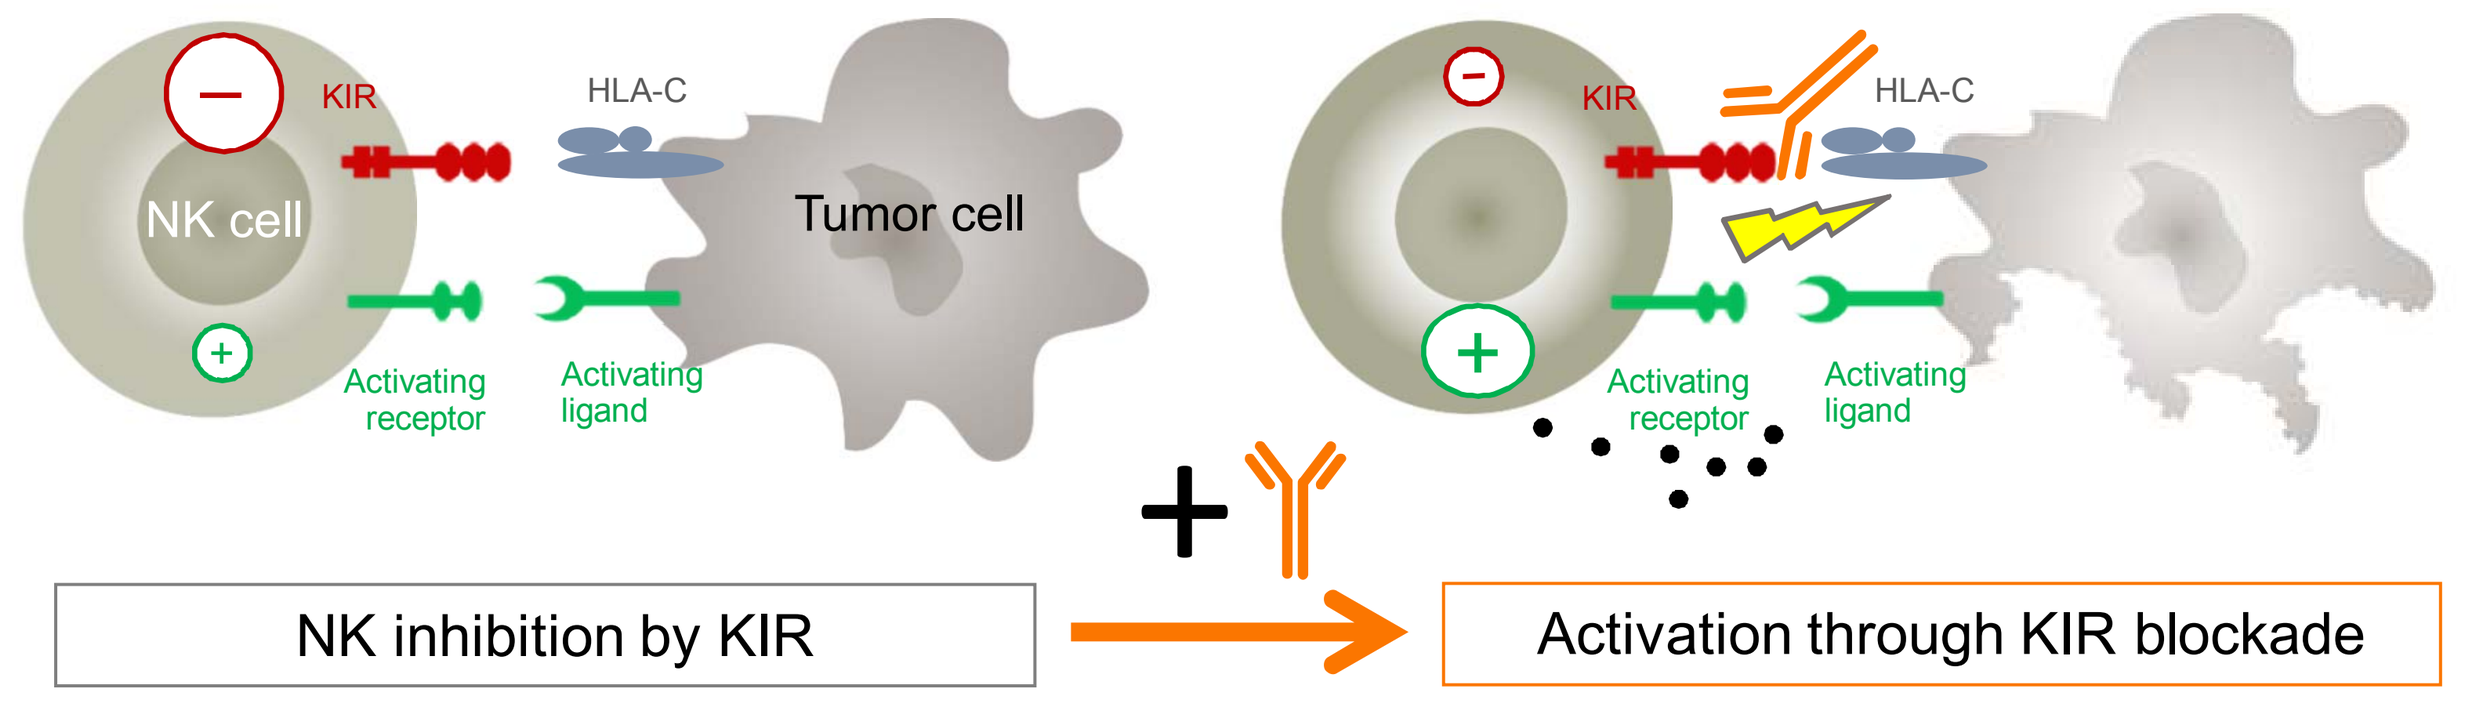
KIR
HLA-C
NK cell
Tumor cell
Activating receptor
Activating ligand
NK inhibition by KIR
Activation through KIR blockade

Blocking NK inhibitory receptor (KIR)

Study Design

The study was designed to evaluate the safety of single agent LIRI and to identify the dose levels resulting, after a single administration, in either sustained full occupancy of KIR for at least 4 consecutive weeks or an intermittent full occupancy of KIR for less than 3 consecutive weeks.

This was an open label, dose-escalation multi-center phase 1 trial in patients with solid tumors (SOL) or hematologic malignancies (HEM). To allow prolonged observation and full pharmacology assessment of LIRI, patients had to have disease in complete response (CR, mandatory in AML), partial response (PR) or slowly progressive (SP).

In the dose escalation 3+3 part (sequential inclusions), six dose levels of LIRI were evaluated: 0.015, 0.3, 1, 3, 6, and 10 mg/kg; for dose levels up to 3 mg/kg, the time between the first 2 infusions was variable, in accordance with the time to KIR desaturation (KIR occupancy < 30%).

For higher dose levels, the duration of cycle 1 was limited to 6 months. Subsequent cycles were administered q4w. Following the dose escalation part of the study, 2 additional cohorts of 8 patients each were entered and treated monthly at 0.015 or 3 mg/kg x 4.

Results

Pretreatment characteristics and exposure to LIRI

	Escalation (n=20)						Extension (n=17)	
dose level	**0.015 mg/kg**	**0.3 mg/kg**	**1 mg/kg**	**3 mg/kg**	**6 mg/kg**	**10 mg/kg**	**0.015 mg/kg**	**3 mg/kg**
HEM	1 CLL, 2 NHL	1 AML, 2 NHL	1 AML, 1 NHL	1 AML, 2 CLL	1 NHL	1 AML, 1 NHL	2 CLL, 2 NHL	1 AML, 1 CLL, 2 NHL
SOL			1 ovarian, 1 other	1 Breast	2 ovarian	1 ovarian	3 breast, 2 ovarian	2 breast, 1 ovarian, 1 other

		AML	CLL	NHL	BREAST	OVARIAN	OTHER	All
Gender	M F	4 (80%) 1 (20%)	5 (83%) 1 (17%)	5 (45%) 6 (55%)	0 6 (100%)	0 7 (100%)	0 2 (100%)	14 (38%) 23 (62%)
Age	Median Range	70.0 62.0;73.0	63.0 40.0;73.0	61.0 53.0;72.0	58.0 33.0;66.0	64.0 64.0;66.0	65.0 36.0;72.0	62.0 33.0;73.0
Disease status	SP PR CR	0 0 5 (100%)	0 5 (83%) 1 (17%)	2 (18%) 5 (45 %) 4 (36%)	0 5 (83%) 1 (17%)	0 4 (57%) 3 (43%)	0 1 (50%) 1 (50%)	2 (5%) 20 (55%) 15 (40%)
Previous systemic therapies	Median Range	1 1;2	1.5 0;7	2 0;5	2.5 2;3	2.0 2;3	 NA	2 0;7
ECOG	0 1	4 (80%) 1 (20%)	3 (50%) 3 (50%)	6 (55%) 5 (45%)	4 (67%) 2 (33%)	4 (57%) 3 (43%)	2 (100%) 0	23 (62%) 14 (38%)

Dose (mg/kg)	No. of patients	Median no. of cycles per patient
0.015	12	4
0.3	3	4
1.0	4	2.5
3.0	12	4
6.0	3	4
10.0	3	1

Treatment emergent adverse events

- No DLT in the dose-escalation; MTD was not reached. Only one SAE (G3 urticaria, at 0.015 mg/kg) was reported as treatment-related.
- Related AEs (>10% of the patients) included fatigue/asthenia (30% of pts), pruritus (20%), infusion related reaction (14%), headache (11%).
- Eighteen patients (49%) discontinued study prematurely: 13 for disease progression, 3 for related AE's (urticaria, elevated LFT, and rash papular) and 2 for other reasons (misreading by site of KIR occupancy and patient convenience).
- Release of cytokines in serum was rare and, whenever it occurred, it was mild to moderate.
- This trial was not designed to detect antitumor as many patients were still responding to prior therapy at study entry.

Patients with treatment-emergent AEs							
Type of AEs	**0.015 mg/kg (n=12)**	**0.3 mg/kg (n=3)**	**1 mg/kg (n=4)**	**3 mg/kg (n=12)**	**6 mg/kg (n=3)**	**10 mg/kg (n=3)**	**Total (n=37)**
Any	11 (91.7%)	3 (100%)	4 (100%)	12 (100%)	3 (100%)	3 (100%)	**36 (97.3%)**
Related	8 (66.7%)	3 (100%)	2 (50%)	9 (75%)	2 (66.7%)	1 (33.3%)	**25 (67.6%)**
Any grade 3 / 4	7 (58.3%)	2 (66.7%)	3 (75%)	9 (75%)	2 (66.7%)	2 (66.7%)	**25 (67.6%)**
Any AE leading to study drug discontinuation	2 (17%)	0	1(25%)	1(8%)	0	2(67)	**6 (16%)**
Any SAE	2 (16.7%) Uriticaria Cholangitis	1 (33.3%) Bacterial sepsis	0	1 (8.3%) HTA	1 (33.3%) Cholangitis	1 (33.3%) Pre-syncope	**6 (16.2%)**

KIR Occupancy and pharmacokinetics

- Full KIR occupancy was sustained during > 4 weeks for dose-levels ≥ 0.3 mg/kg.
- LIRI concentration versus time profile shows an initial distribution phase followed by a long terminal elimination phase classical for mAb, and suggests dose dependent linear PK at high doses (from 0.3 mg/kg), with low to moderate interpatient variability.
- Exploratory covariate analysis showed potential effect of body weight, sex, baseline NK and T cell number, %KIR on T cell number on PK .
- Only one patient at 0.015mg/kg in the extension phase became significantly positive (titer=3) for HAHA from cycle 2.

KIR Occupancy

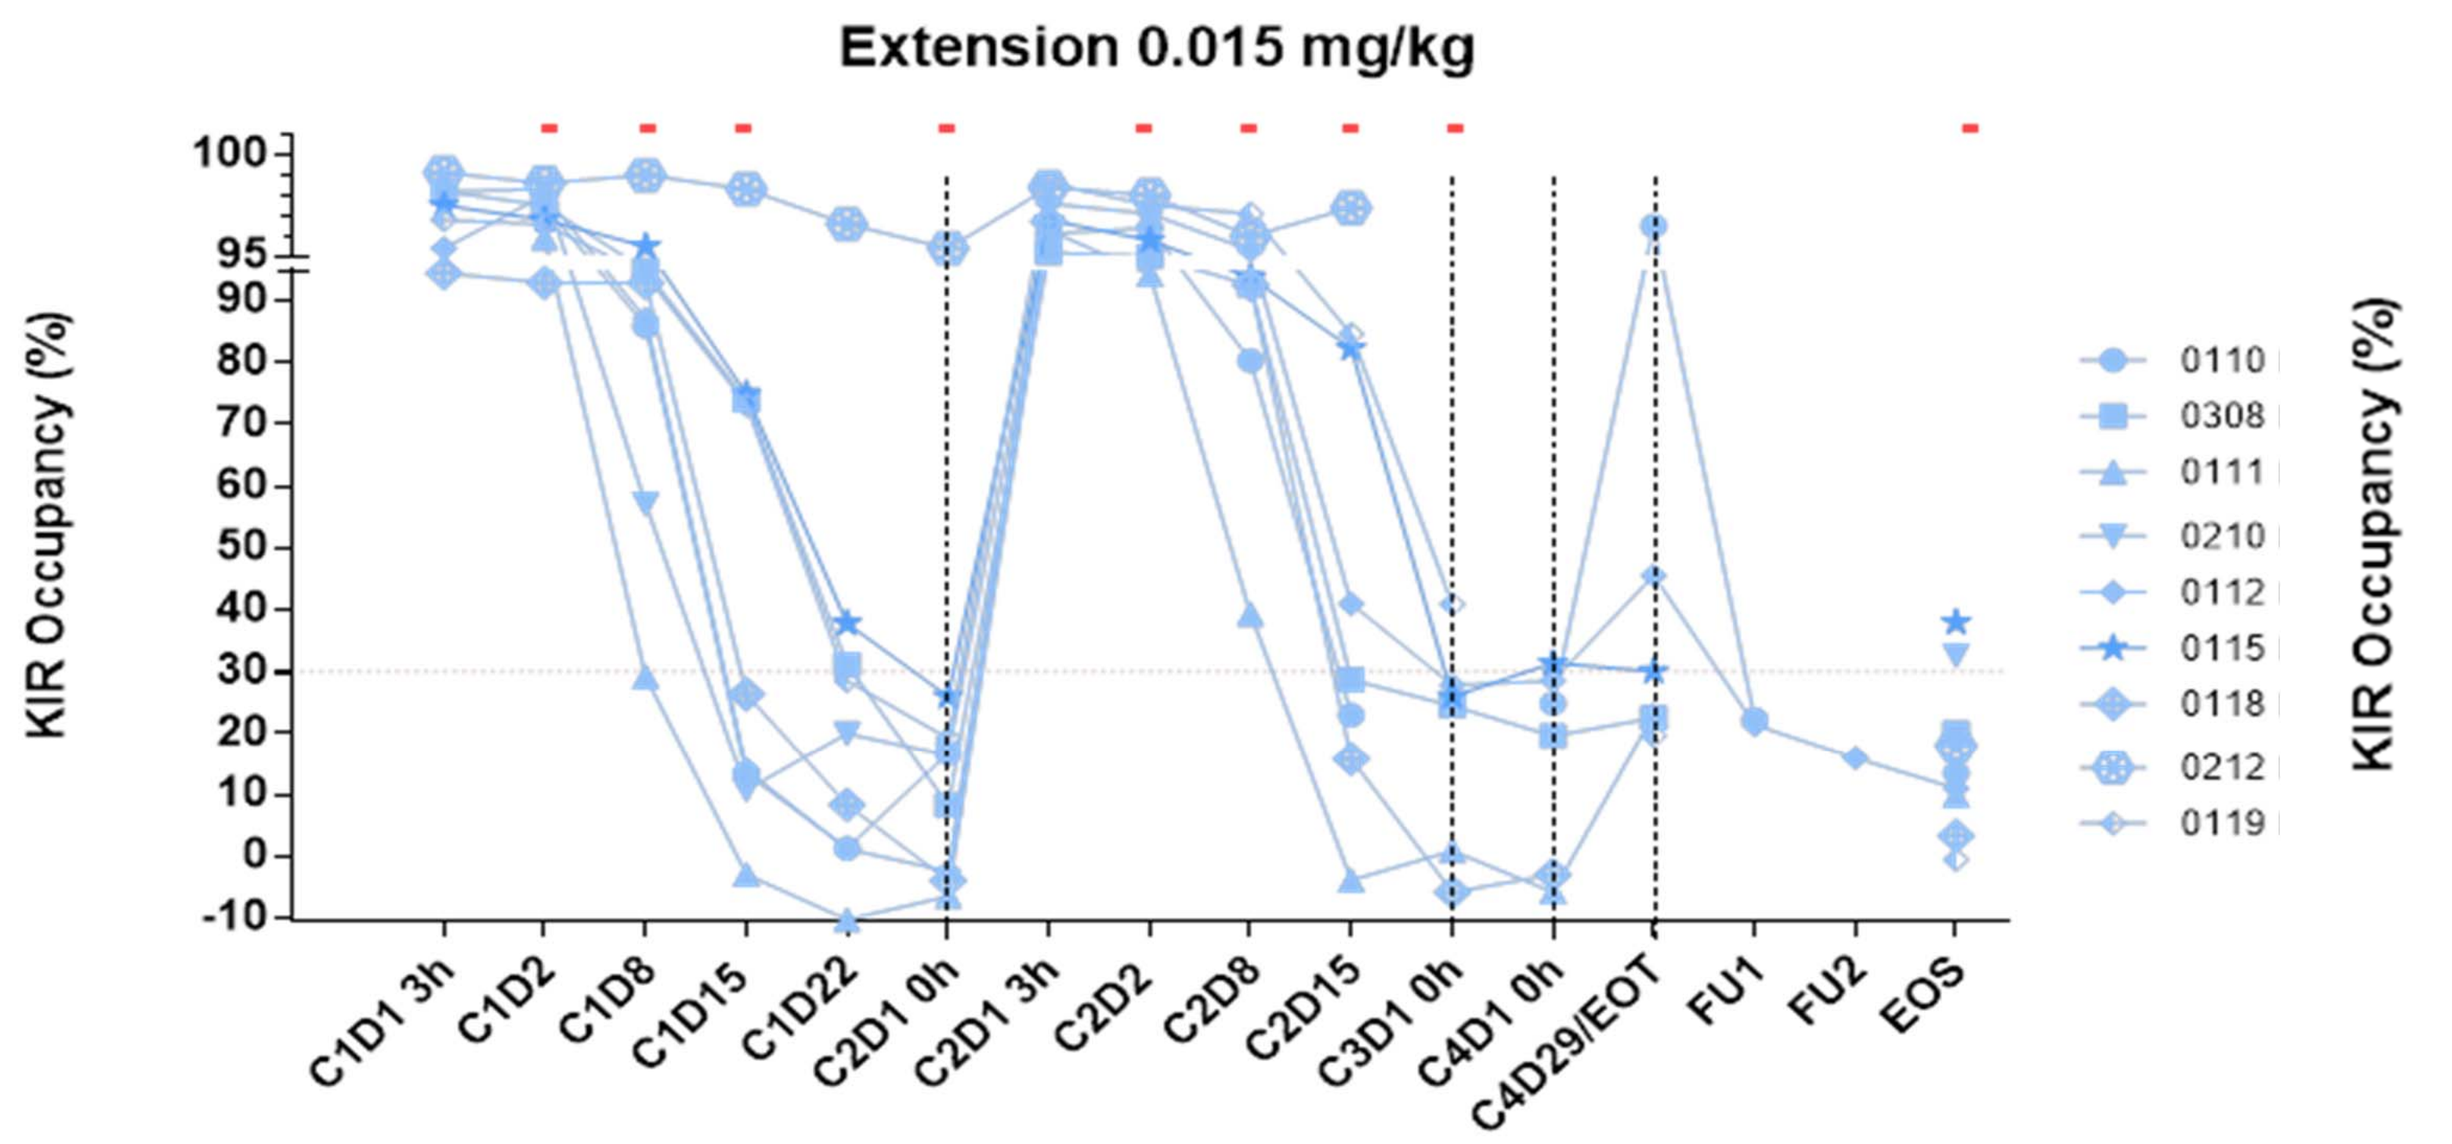
Extension 0.015 mg/kg
KIR Occupancy (%)
C1D1 3h
C1D2
C1D8
C1D15
C1D22
C2D1 0h
C2D1 3h
C2D2
C2D8
C2D15
C3D1 0h
C4D1 0h
C4D29/EOT
FU1
FU2
EOS
0110
0308
0111
0210
0112
0115
0118
0212
0119

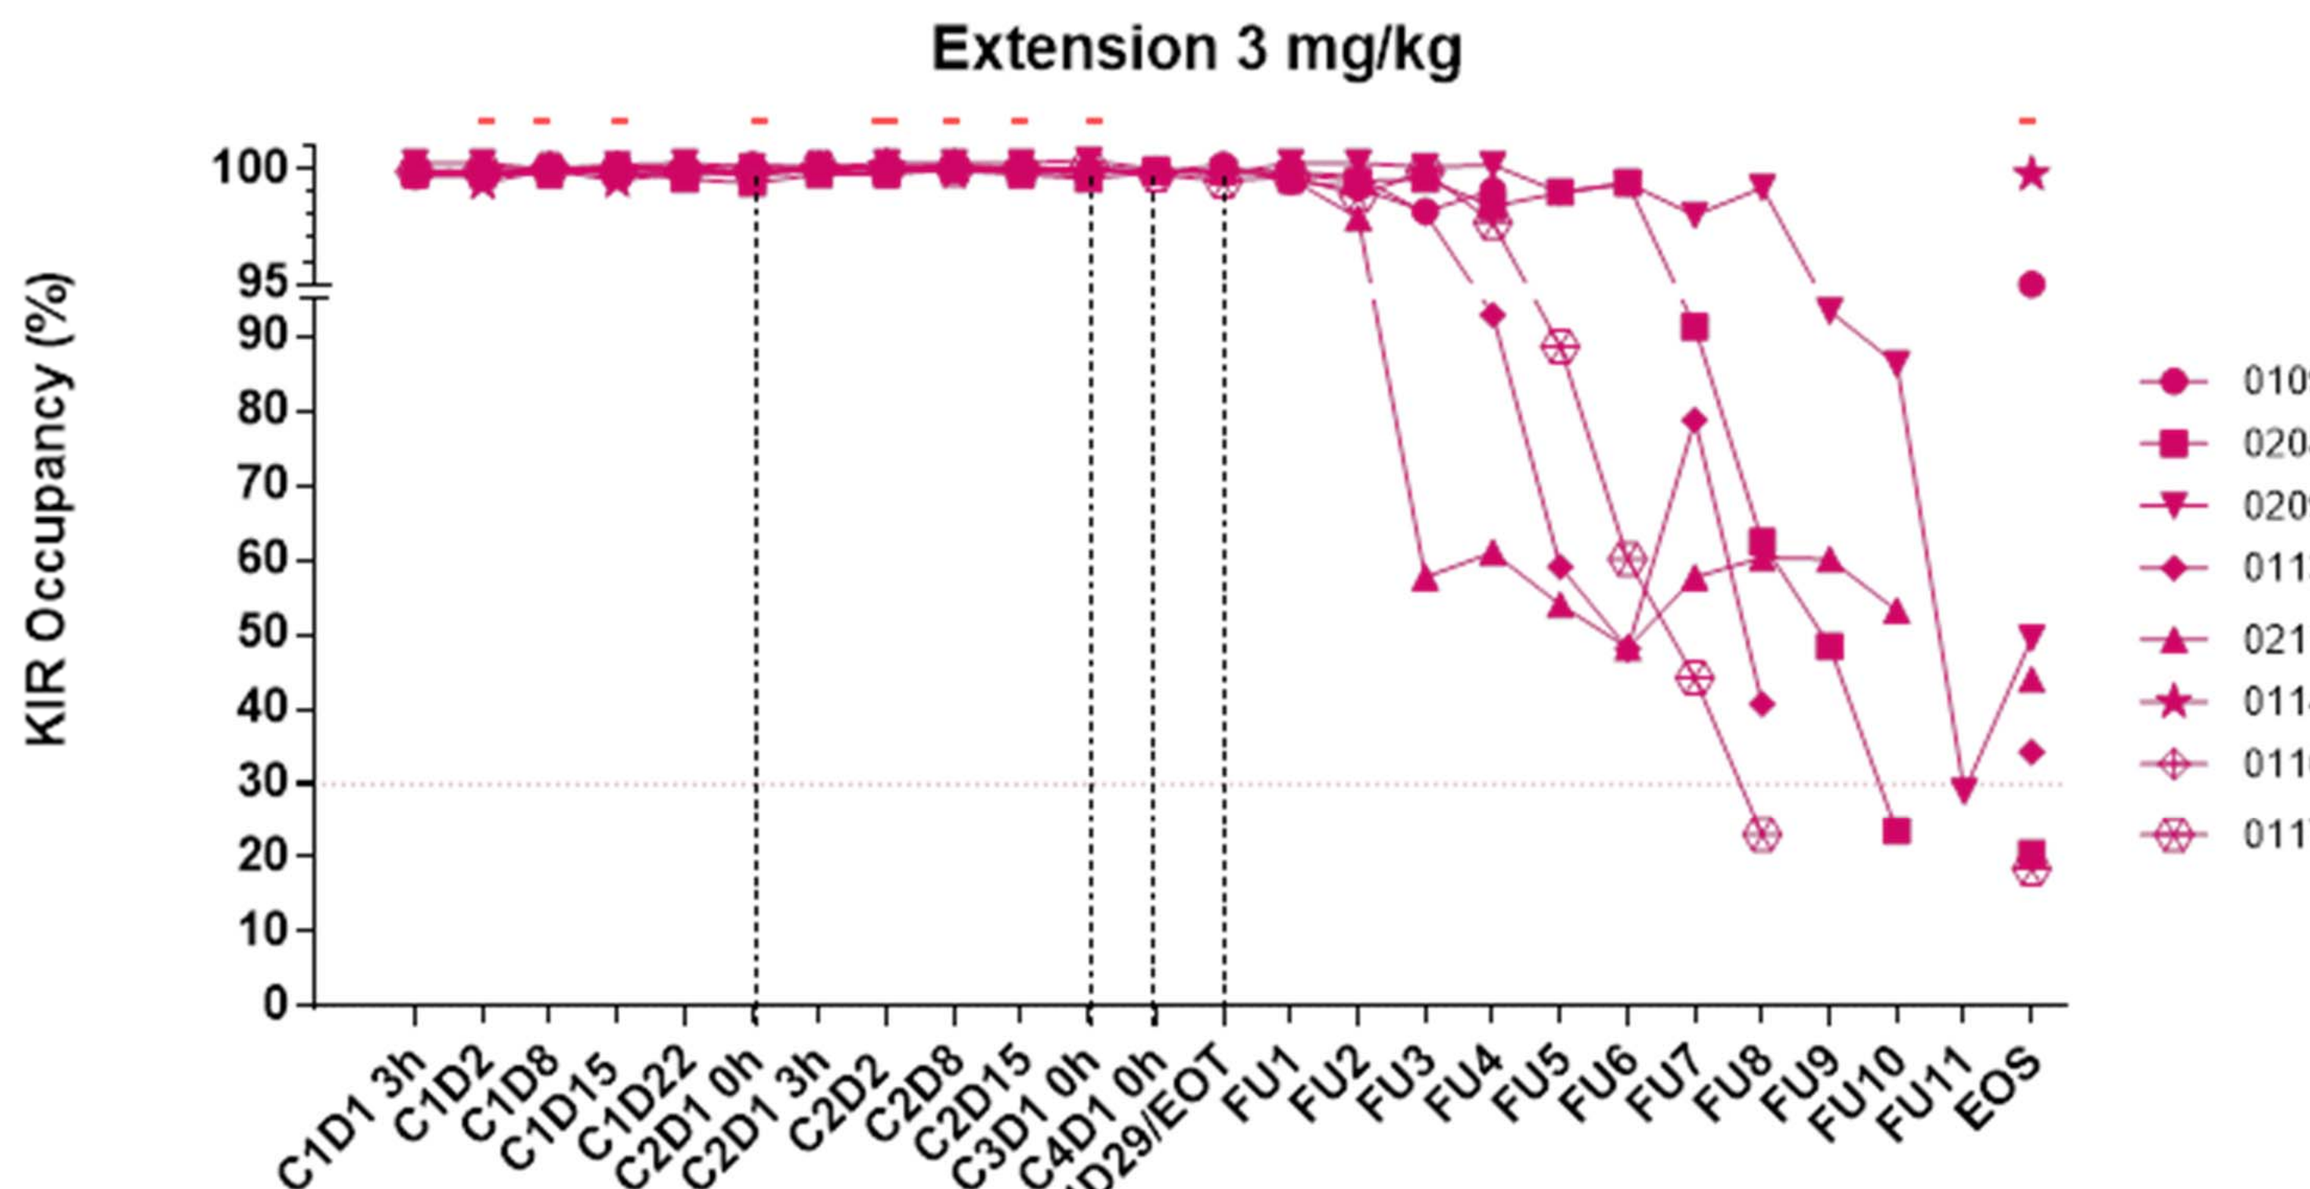
Extension 3 mg/kg
KIR Occupancy (%)
0109
0208
0209
0113
0211
0114
0116
0117

Pharmacokinetics

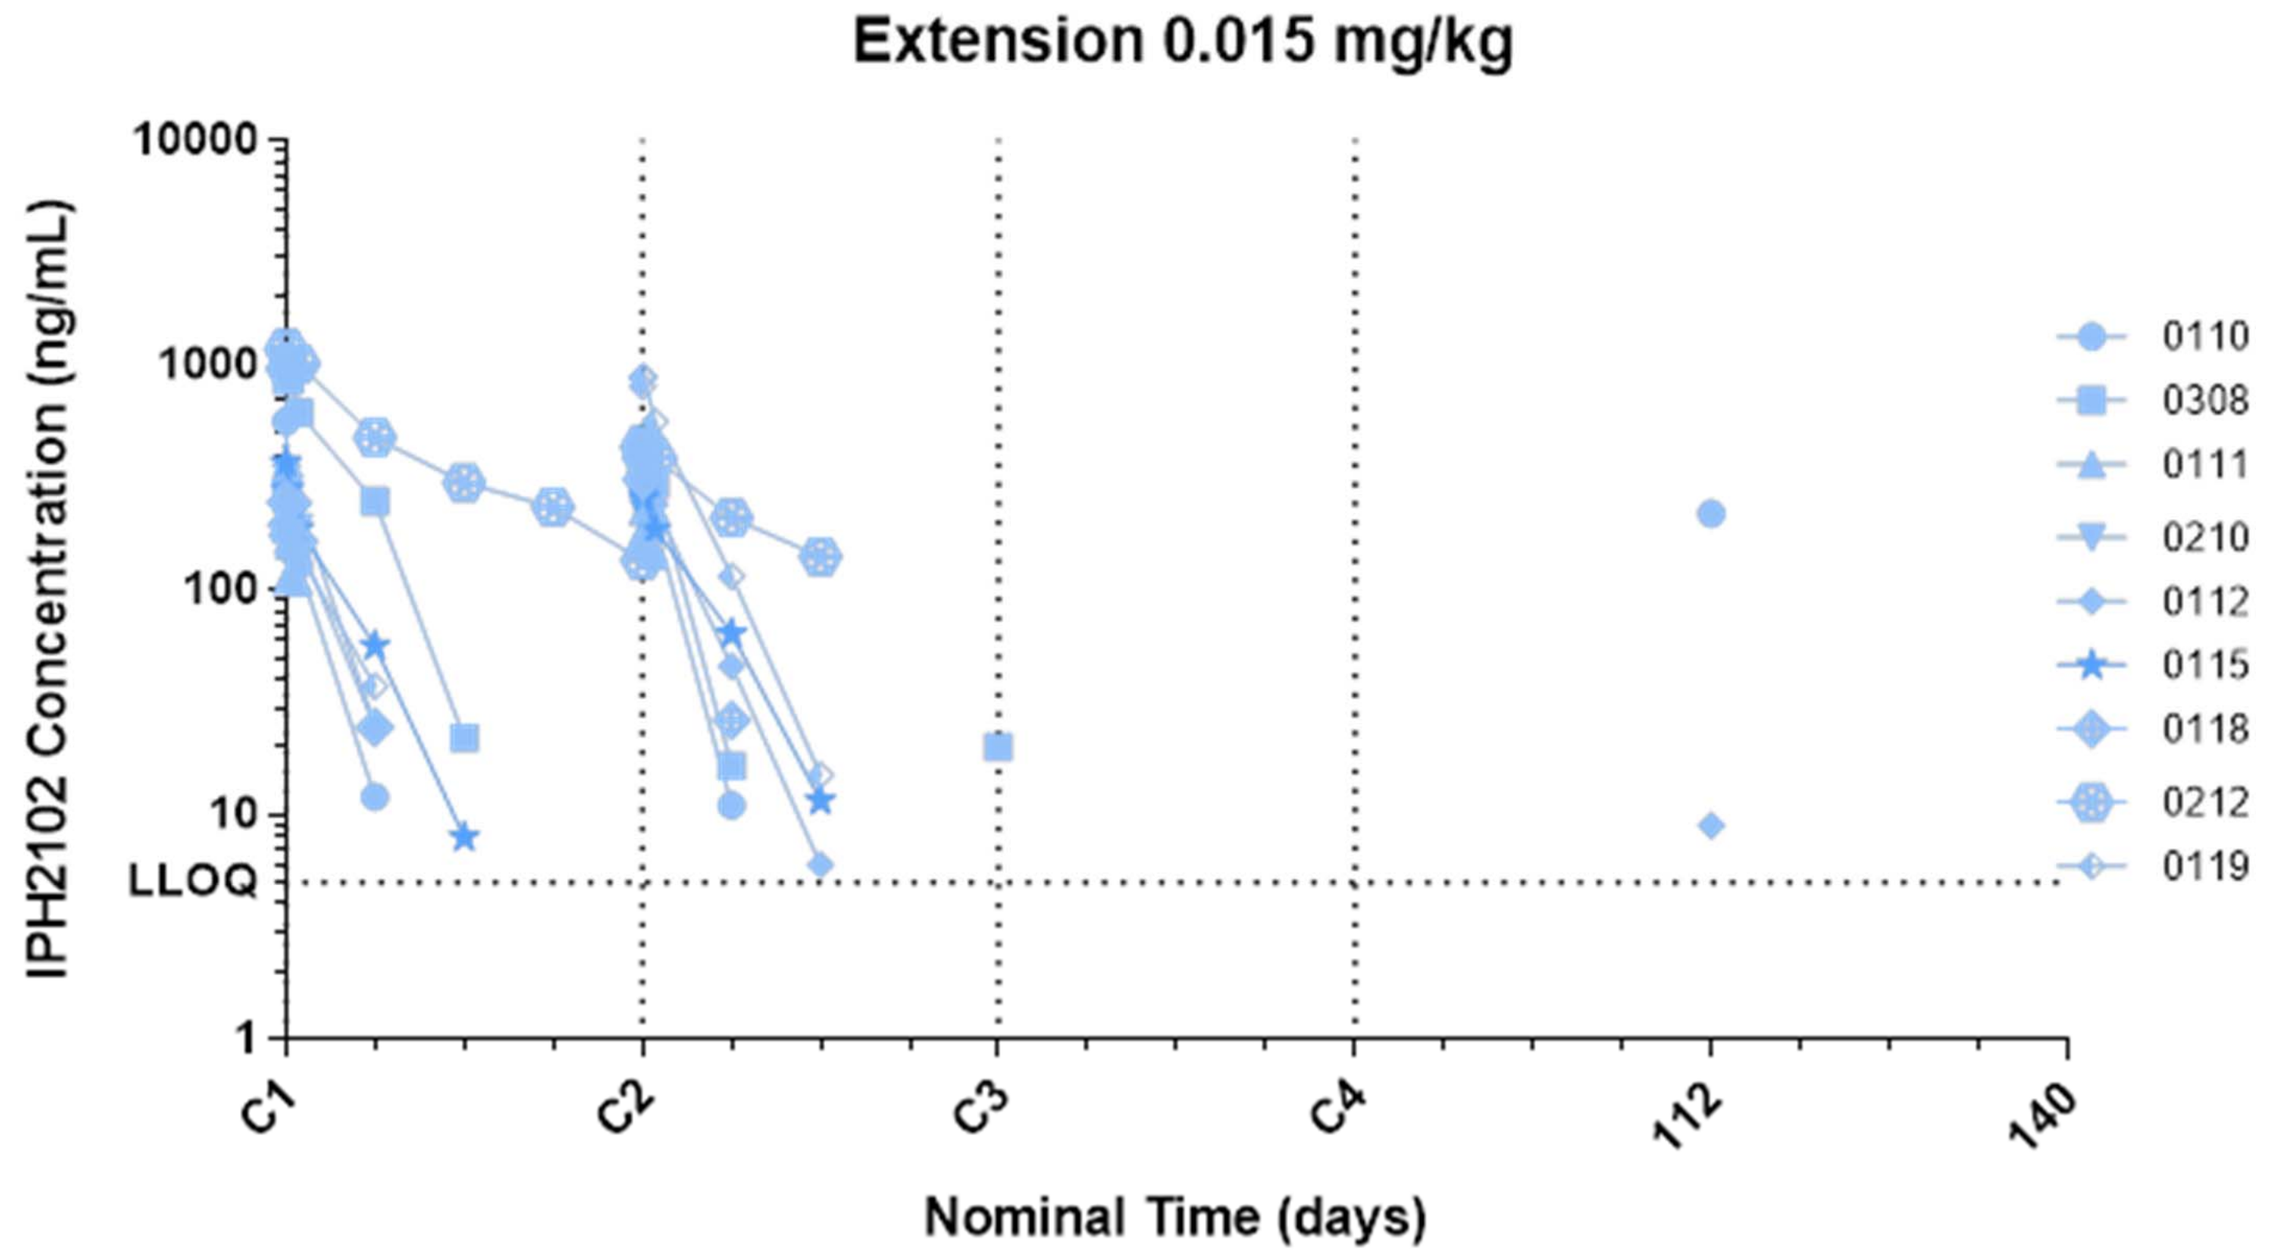
Extension 0.015 mg/kg
IPH2102 Concentration (ng/mL)
LLOQ
Nominal Time (days)
0110
0308
0111
0210
0112
0115
0118
0212
0119

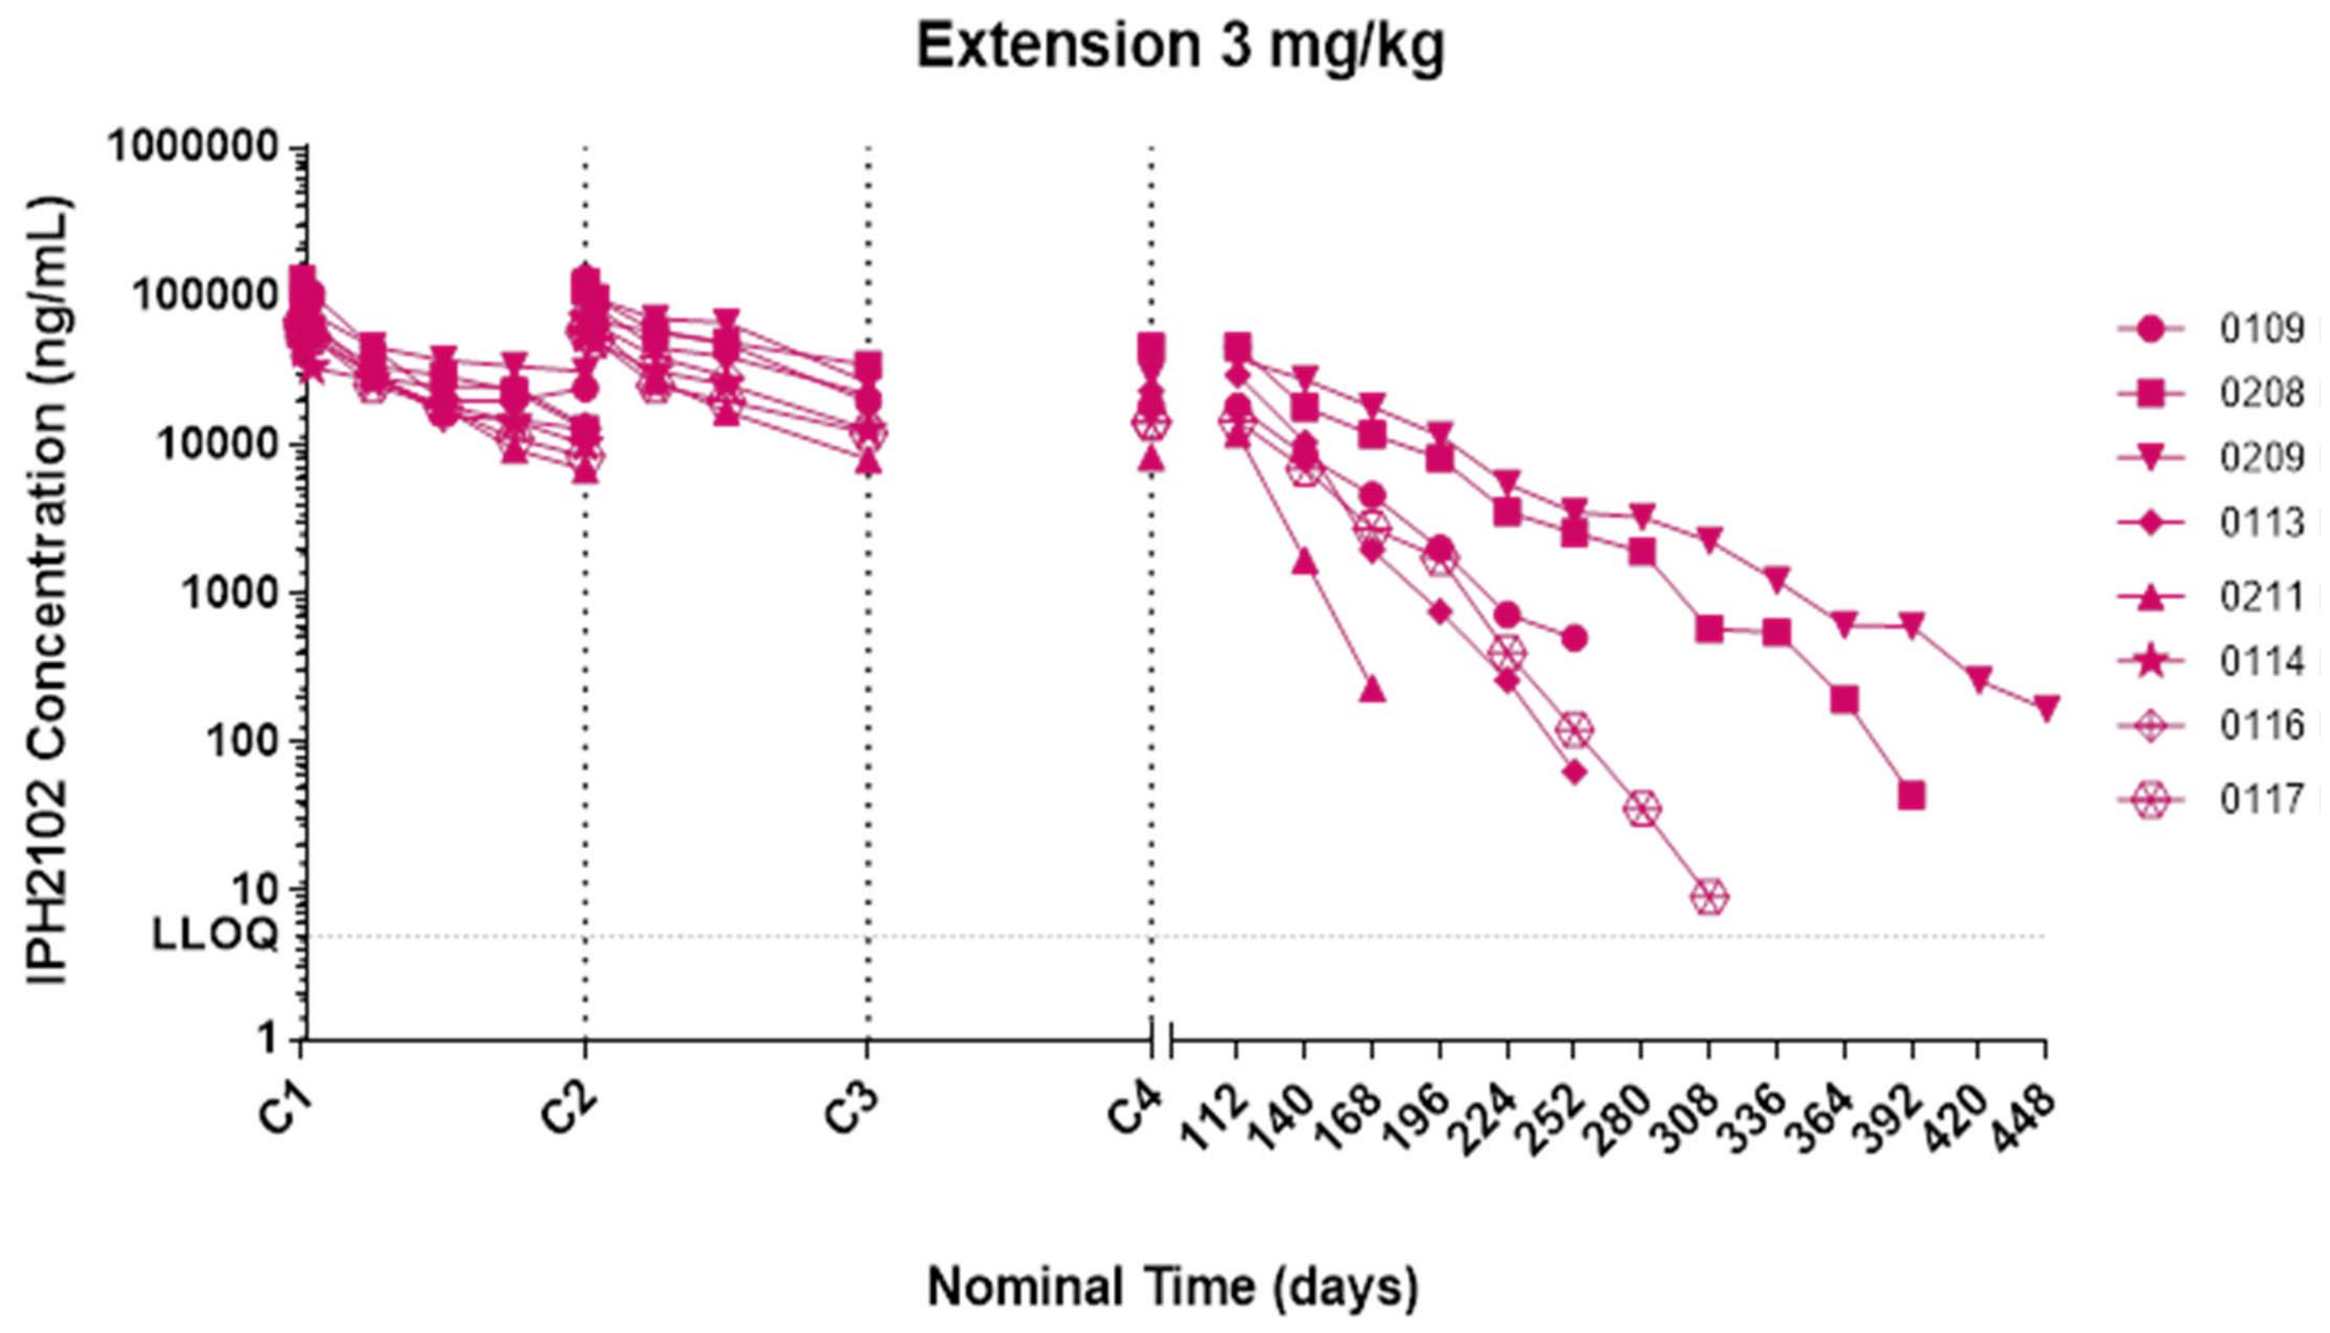
Extension 3 mg/kg
IPH2102 Concentration (ng/mL)
LLOQ
Nominal Time (days)
0109
0208
0209
0113
0211
0114
0116
0117

Conclusions

LIRI was generally well tolerated up to the highest dose tested of 10 mg/kg which was associated with prolonged KIR occupancy for at least 6 months.

Few transient, usually mild to moderate infusion related, skin reactions and one possibly related grade 2 allergy were the only immune related disorders observed in this trial.

LIRI is currently investigated in a randomized phase 2 trial as maintenance therapy for elderly patients with AML in CR1 and in various phase 1 combination studies in solid tumors (nivolumab, ipilimumab) and hematologic malignancies (nivolumab, elotuzumab, vidaza).

References

(1) Caligiuri MA et al Hematology Am Soc Hematol Educ Program. 2004:337-53
(2) Ruggeri L et al. Blood 2007 110(1):433-40
(3) Vey N et al. Blood 2012; 120(22): 4317-23
(4) Benson DM et al. Blood 2012; 120(22): 4324-33

innate pharma

ASCO 2015